Antibody Fc Engineering for Optimal Engagement of Innate Immune Cells

John Richards, Greg Lazar, Sher Karki, Sreenivas Laxmanan, Aaron Chamberlain, & John Desjarlais

Xencor, Inc. 111 W. Lemon Ave., Monrovia, CA 91016

Proteins by Design®

Abstract

Antibody Fc gamma receptors (FcγRs) are present on a wide variety of effector cell populations, including NK cells, dendritic cells (DC), neutrophils, and macrophages. We have previously engineered the Fc domains of antibodies to enhance their affinity for FcγRIIIa, leading to dramatic enhancements in ADCC. While NK cell-mediated lysis occurs predominantly through a single activating receptor FcγRIIIa, activation of antigen presenting cells such as DC and macrophages can also be influenced by the activating receptor FcγRIIa and inhibitory receptor FcγRIIb. In an effort to optimally tune antibodies for effector function, we have engineered a panel of Fc variants with a variety of unique FcγR affinities and specificities, including selective engagement of FcγRIIIa and FcγRIIa over FcγRIIb. Results indicate that whereas NK cell-mediated killing is correlated strongly with FcγRIIIa affinity, phagocytosis by macrophages is dependent on binding to both FcγRIIa and FcγRIIIa. Surprisingly, our data show that absolute FcγRIIb affinity does not negatively impact phagocytosis. Overall these variants have the potential to improve anti-cancer therapy by increasing not only innate effector functions, but also by enhancing adaptive anti-tumor responses, a novel feature of engineered therapeutic antibodies.

Effector cells, effector functions, and activating vs. inhibitory FcγRs

Cell type	Anti-tumor relevance		FcγR-dependent function(s)	FcγR expression
	colon, breast, lung, ovarian	lymphomas		
NK	low?	high?	ADCC	IIIa
Neutrophil	?	high?	Phagocytosis ADCC	[I, IIa] vs IIb, IIIb
Macrophage	high?	?	Phagocytosis ADCC	[I, IIa, IIIa] vs [IIb] [I, IIa, IIIa] vs [IIb]
DC	high?	?	Phagocytosis Antigen presentation	IIa vs IIb

Literature supporting relevance of FcγRIIa and monocyte-derived effector cells

- Higher FcγRIIa affinity leads to longer progression free survival in anti-GD2 neuroblastoma therapy (Cheung et al., JCO 2006)
- FcγRIIa-mediated phagocytosis by human DCs leads to enhanced DC maturation and CTL priming (Dhodapkar et al., J Exp Med 2002; Groh et al., PNAS 2005; Boruchov et al., J Clin Invest 2005)
- Antibody efficacy in murine models
 - Dependent on monocyte population (Hamaguchi et al., J Exp Med 2006)
 - Dependent on FcγRIV, expressed on monocytes/macrophages (Nimmerjahn & Ravetch, Science 2005)
 - Dependent on T cells (Dyall et al., Eur J Immunol 1999; Wolpoe et al., J Immunol 2003)

FcγR-mediated effector functions

Lysis, Apoptosis

Phagocytosis
Antigen presentation

Secondary effects

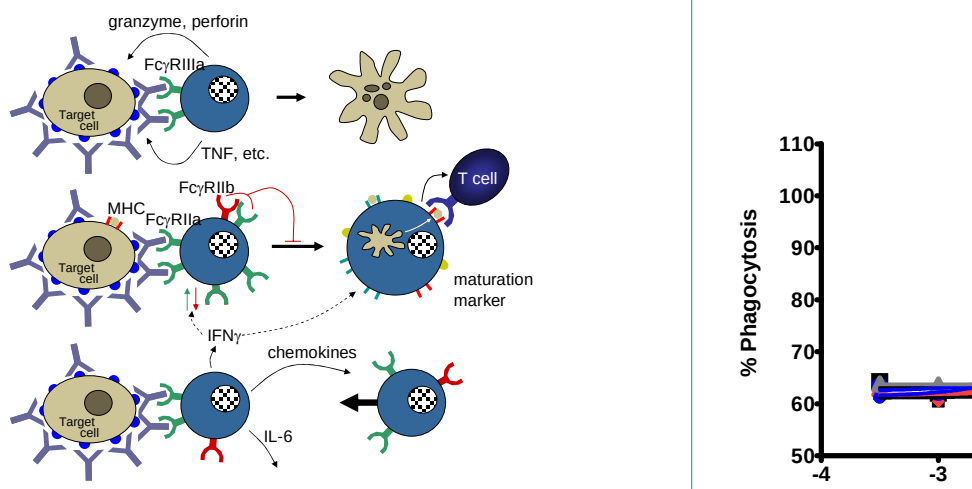
granzyme, perforin
FcγRIIIa
Target cell
TNF, etc.
FcγRIIb
MHC
FcγRIIa
T cell
maturation marker
IFNγ
chemokines
IL-6

A novel S2 substitution selectively enhances FcγRIIa affinity

BIAcore sensorgrams for R131 FcγRIIa

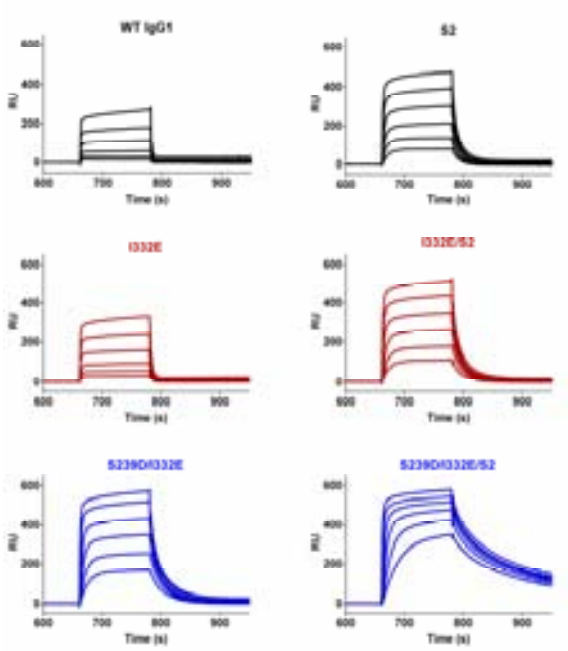
WT IgG1
S2
I332E
I332E/S2
S239D/I332E
S239D/I332E/S2
RU
Time (s)

Anti-EpCAM mAbs
(humanized 17-1A)

His-tagged human FcγR
extracellular domains

BIAcore:
Immobilized antibody
FcγR analyte at multiple concentrations
Global Langmuir fitting

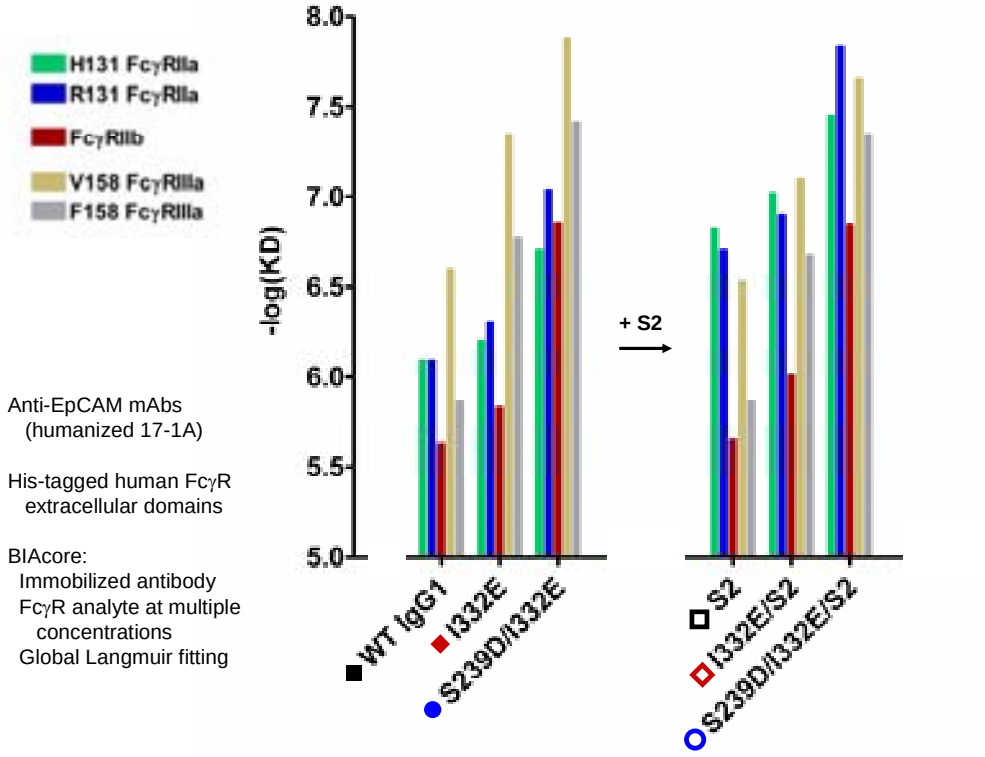
H131 FcγRIIa
R131 FcγRIIa
FcγRIIb
V158 FcγRIIIa
F158 FcγRIIIa
-log(KD)
8.0
7.5
7.0
6.5
6.0
5.5
5.0
+ S2
WT IgG1
I332E
S239D/I332E
S2
I332E/S2
S239D/I332E/S2

FcγRI –log(KD)'s: WT IgG1(9.05) I332E(8.88) S239D/I332E(9.30) S2(8.09) I332E/S2(9.00) S239D/I332E/S2(8.90)

Defucosylation affects only FcγRIIIa

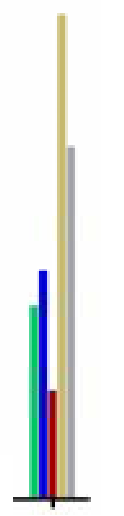

WT IgG1
— Fucose
(by Lec13 expression)

Antibody-mediated cytokine release correlates with FcγR affinity

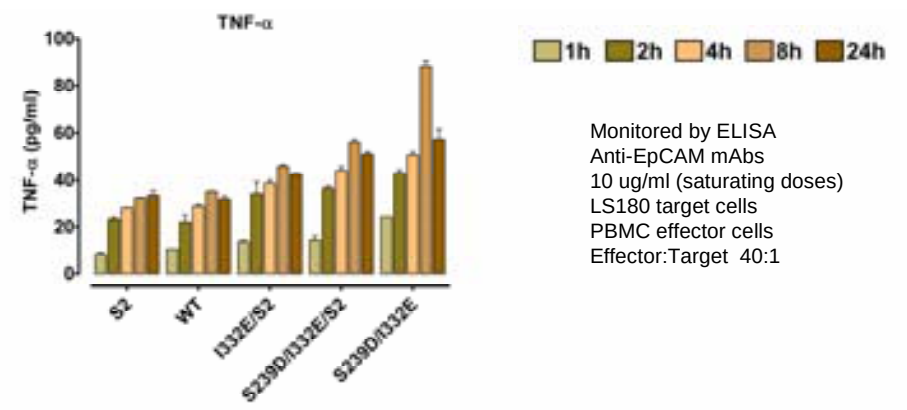
TNF-α
1h
2h
4h
8h
24h
TNF-α (pg/ml)
S2
WT
I332E/S2
S239D/I332E/S2
S239D/I332E

Monitored by ELISA
Anti-EpCAM mAbs
10 ug/ml (saturating doses)
LS180 target cells
PBMC effector cells
Effector:Target 40:1

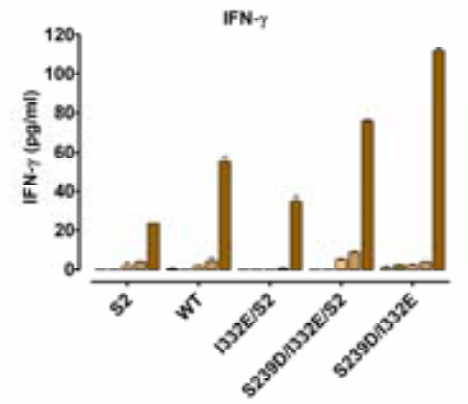
IFN-γ
IFN-γ (pg/ml)
S2
WT
I332E/S2
S239D/I332E/S2
S239D/I332E

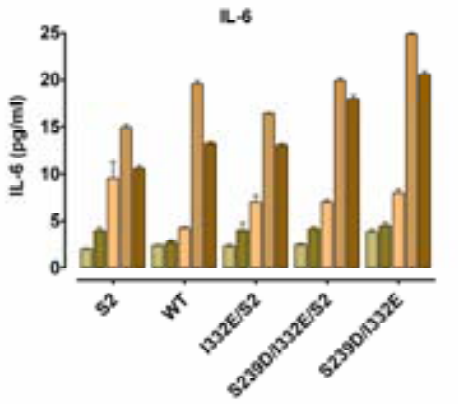
IL-6
IL-6 (pg/ml)
S2
WT
I332E/S2
S239D/I332E/S2
S239D/I332E

increasing FcγR affinity

Flow Cytometric Readout of Phagocytosis Assay

WT IgG1

S239D/I332E

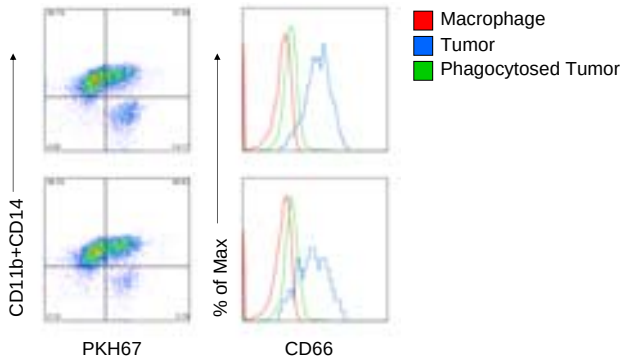
CD11b+CD14
PKH67
% of Max
CD66
Macrophage
Tumor
Phagocytosed Tumor

Phagocytosis by macrophages is enhanced by S2 and other Fc substitutions against lymphoma cell lines

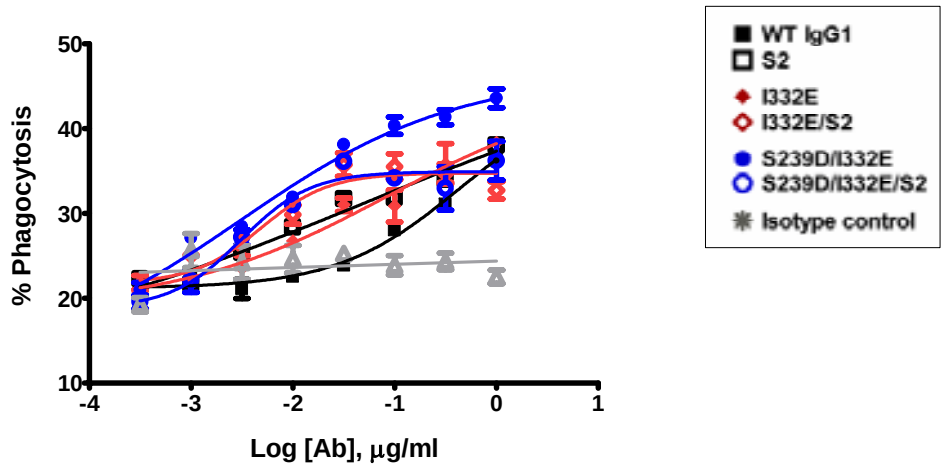
% Phagocytosis
Log [Ab], μg/ml
WT IgG1
S2
I332E
I332E/S2
S239D/I332E
S239D/I332E/S2
Isotype control

Monitored by cytometry
Anti-CD19 mAbs
Raji target cells
MDM effector cells
Donor genotype: F/F, R/H
Effector:Target: 3:1
1% Human Serum

Cytometry of these MDM (not shown) indicated expression levels:

FcgRI	83 %
FcgRIIa/IIb	98 %
FcgRIIIa	49 %

Phagocytosis by macrophages is enhanced by S2 and other Fc substitutions against colon cancer cell lines

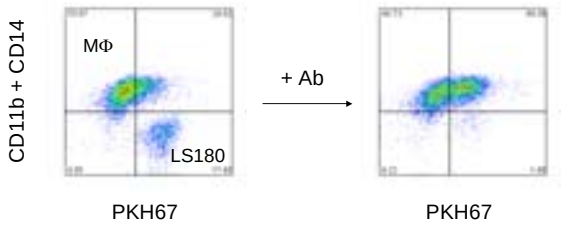
CD11b + CD14
MΦ
LS180
PKH67
+ Ab
PKH67

Monitored by cytometry
Anti-EpCAM mAbs
LS180 target cells
MDM effector cells
Donor genotype: F/F, R/R
Effector:Target: 4:1
1% Human Serum

Cytometry of these MDM (not shown) indicated expression levels:

FcγRI	43 %
FcγRIIa/IIb	95 %
FcγRIIIa	51 %

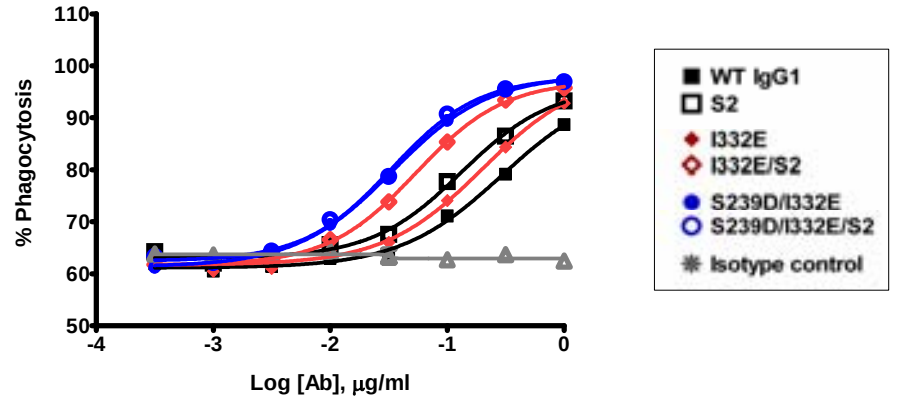
% Phagocytosis
Log [Ab], μg/ml
WT IgG1
S2
I332E
I332E/S2
S239D/I332E
S239D/I332E/S2
Isotype control

Defucosylation has no effect on phagocytosis, however, Fc Engineering can enhance it.

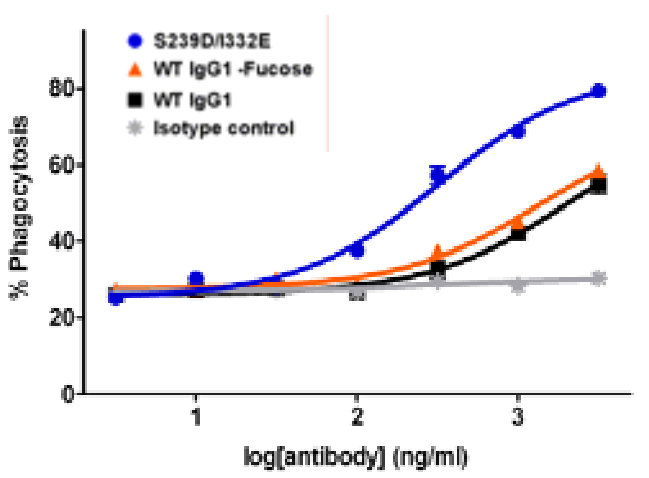
S239D/I332E
WT IgG1 -Fucose
WT IgG1
Isotype control
% Phagocytosis
log[antibody] (ng/ml)

- Defucosylated mAb produced using Lec13
- Anti-EpCAM mAbs
- LS180 target cells
- MDM effector cells

Summary score sheet for variant activities

(Anti-EpCAM, LS180 target cells)

Cell type	FcγR	FcγR-dependent function(s)	S239D/ I332E	I332E	WT	S2	I332E/ S2	S239D/ I332E/ S2
PBMC	all	ADCC	+++	++	+	-	+	+++
		Cytokine release	+++	++	+	-	+	++
NK	IIIa	ADCC	+++	++	+	-	+	+++
Macrophage	I, IIa, IIIa IIb	Phagocytosis	+++	++	+	++	+++	+++
		Cytokine release	in progress					
Neutrophil	I, IIa, IIIa IIb	Phagocytosis						
		ADCC						
moDC	IIa IIb	Maturation						
		Cytokine release						
Mutations			2	1	0	1	2	3

Summary

- Engineered Fc variants enhance a variety of FcγR-mediated effector functions
 - Enhanced ADCC by PBMC and/or NK cells
 - Enhanced phagocytosis by macrophages
 - Solid Tumors
 - Liquid Tumors
 - Enhanced cytokine release by PBMC
- A novel variant, S2, selectively increases FcγRIIa affinity in multiple contexts
- Combination variants have broadly enhanced FcγR activities

Acknowledgements

We thank the following people for helpful discussions and technical assistance:

Bassil Dahiyat, Lori Kunkel, Phil Hammond, Eugene Zhukovsky, Wei Dang, Seung Chu, Hsing Chen, Anjali Simh

Madhav Dhodapkar, Lou Weiner, Raphael Clynes

© 2007 Xencor, Inc. All Rights Reserved.

jrichards@xencor.com www.xencor.com