Coengagement of BCR coreceptor CD19 and inhibitory Fc receptor γRIIb suppresses B cell function: a potential therapeutic strategy for autoimmune disease

Seung Y. Chu, Igor Vostiar, Sher Karki, Gregory L. Moore, Greg A. Lazar, Erik Pong, Hsing Chen, John Richards, Araz Eivazi, Patrick Joyce, Ethan Li, Shaotang Ren, Chi-Ming Yu, David E. Szymkowski*, John R. Desjarlais

Xencor

Introduction

- Depending on the stage of B cell differentiation and the local environment, B cell antigen receptor (BCR) engagement by antigen or immune complex can induce B cell proliferation, apoptosis, or anergy.
- Coengagement of the B cell inhibitory receptor FcγRIIb with the BCR complex via a dual-targeting antibody acts as a surrogate for FcγRIIb downmodulation of the BCR, and may represent a potential therapeutic strategy to suppress autoimmune disease.
- We used antibody engineering combined with Fcγ receptor binding and B cell activation screening assays to identify novel anti-CD19 antibodies containing Fc domains binding with high affinity and selectivity for FcγRIIb relative to the activating receptors FcγRI, FcγRIIa, and FcγRIIIa.

A Inhibitory receptor FcγRIIb feedback-regulates BCR activation of B cells

1. Normal BCR activation leads to calcium mobilization and B cell viability

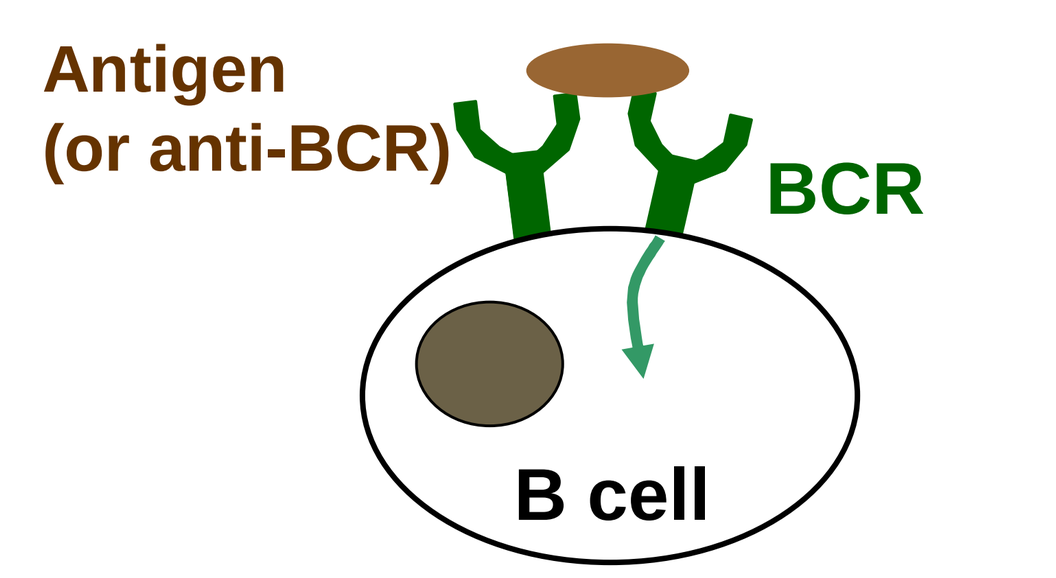
Antigen
(or anti-BCR)
BCR
B cell

BCR-dependent calcium mobilization

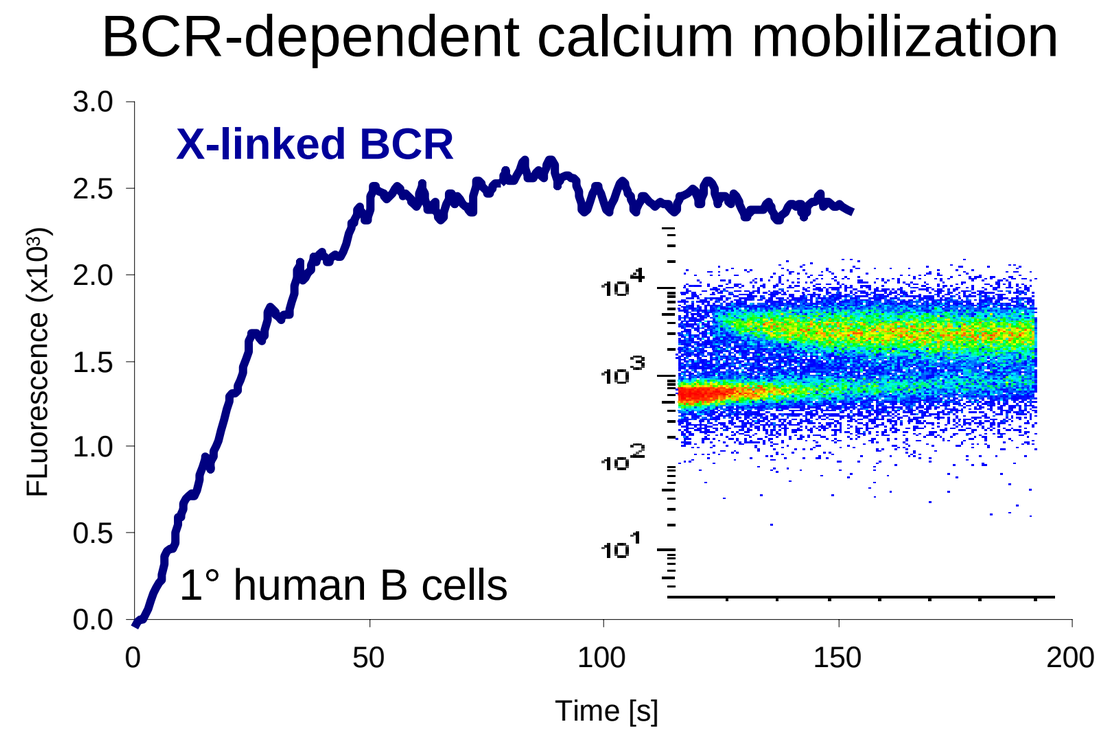
X-linked BCR
1° human B cells
FLuorescence (x10³)
Time [s]

BCR-dependent B cell proliferation

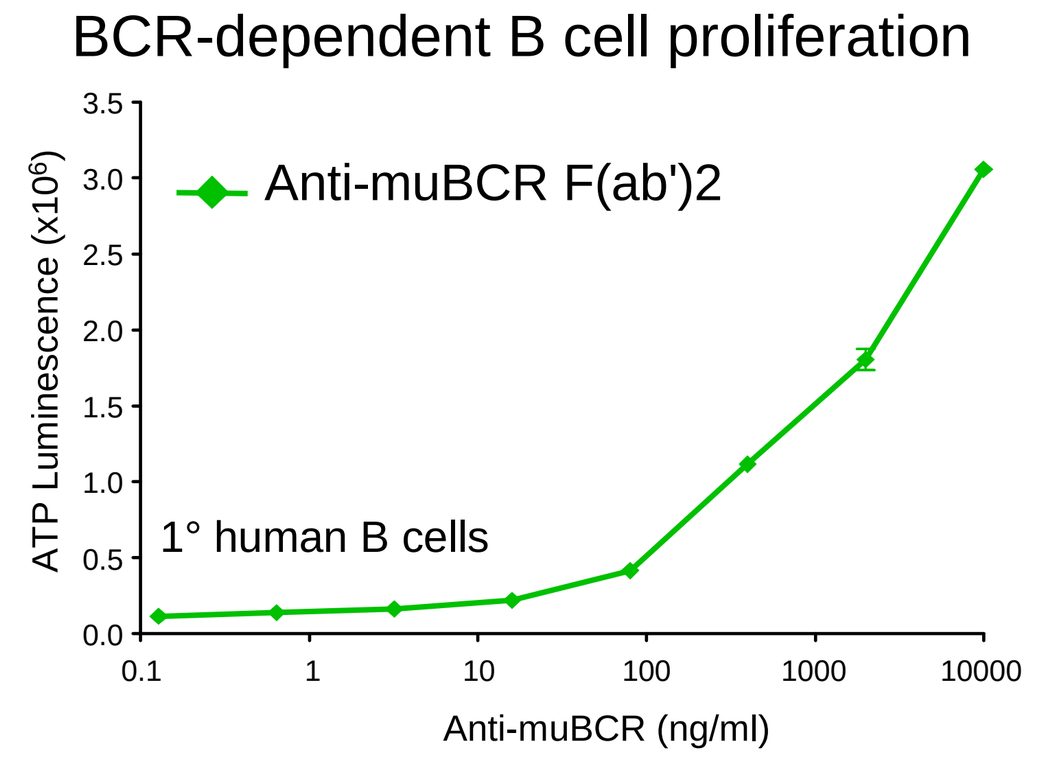
Anti-muBCR F(ab')2
1° human B cells
ATP Luminescence (x10⁶)
Anti-muBCR (ng/ml)

2. B cell activity is downmodulated via inhibitory IgG Fc receptor FcγRIIb

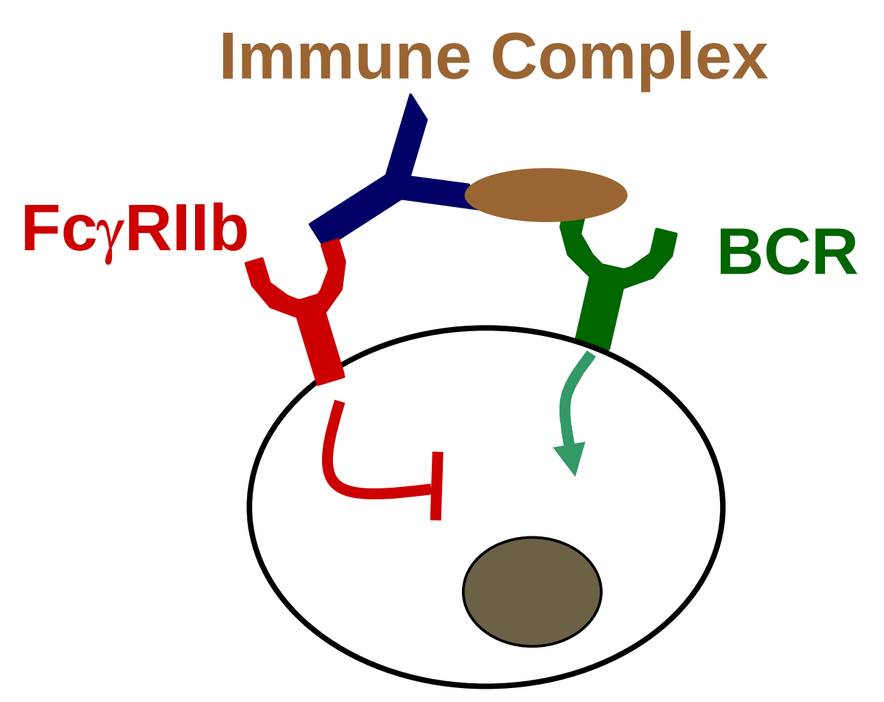
Immune Complex
FcγRIIb
BCR

BCR activation/FcγRIIb inhibition assay

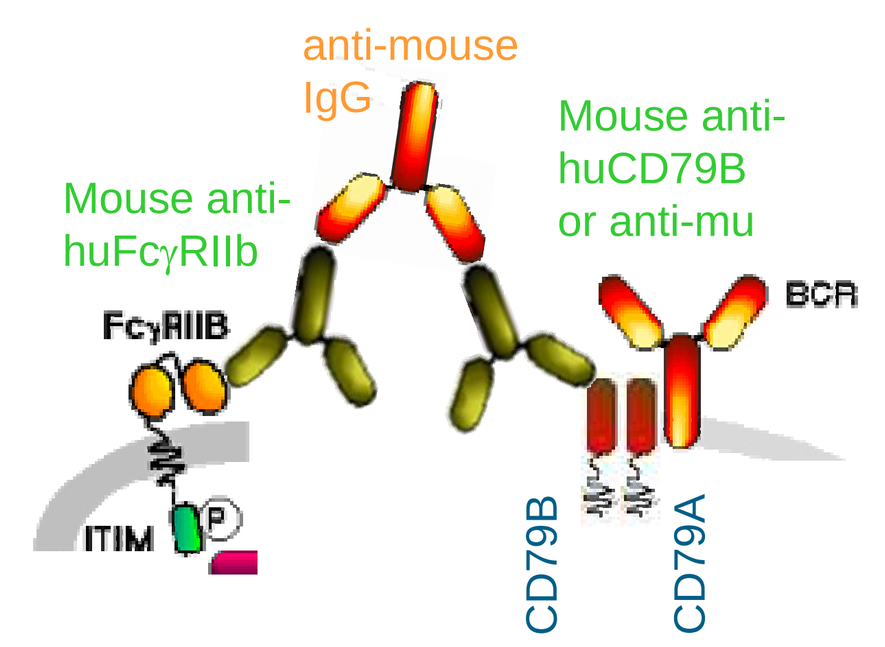
anti-mouse IgG
Mouse anti-huFcγRIIb
Mouse anti-huCD79B or anti-mu
FcγRIIB
BCR
ITIM
CD79B
CD79A

Calcium mobilization inhibited by anti-FcγRIIb

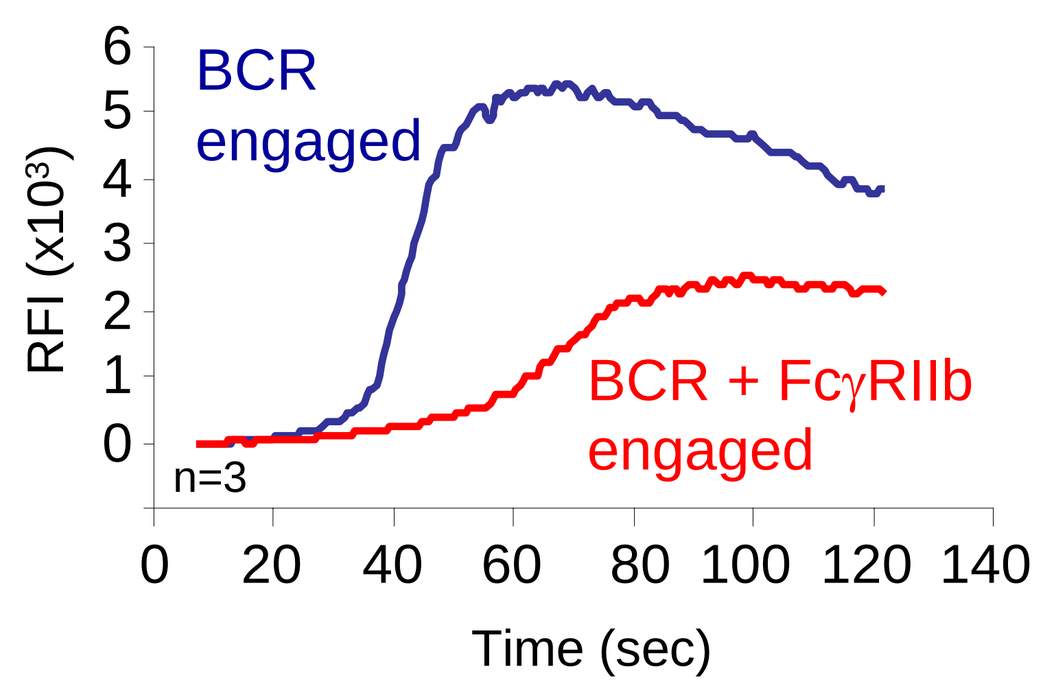
BCR engaged
BCR + FcγRIIb engaged
n=3
RFI (x10³)
Time (sec)

B cell viability inhibited by anti-FcγRIIb

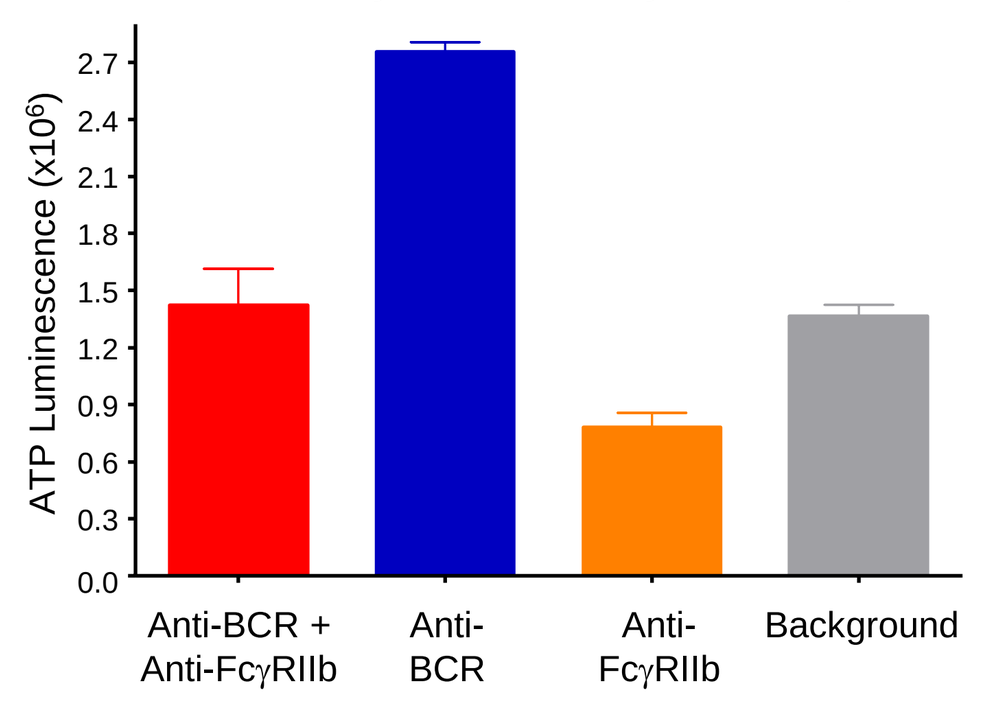
ATP Luminescence (x10⁶)
Anti-BCR + Anti-FcγRIIb
Anti-BCR
Anti-FcγRIIb
Background

3. Direct coengagement of FcγRIIb and BCR coreceptor may be a broadly applicable therapeutic approach

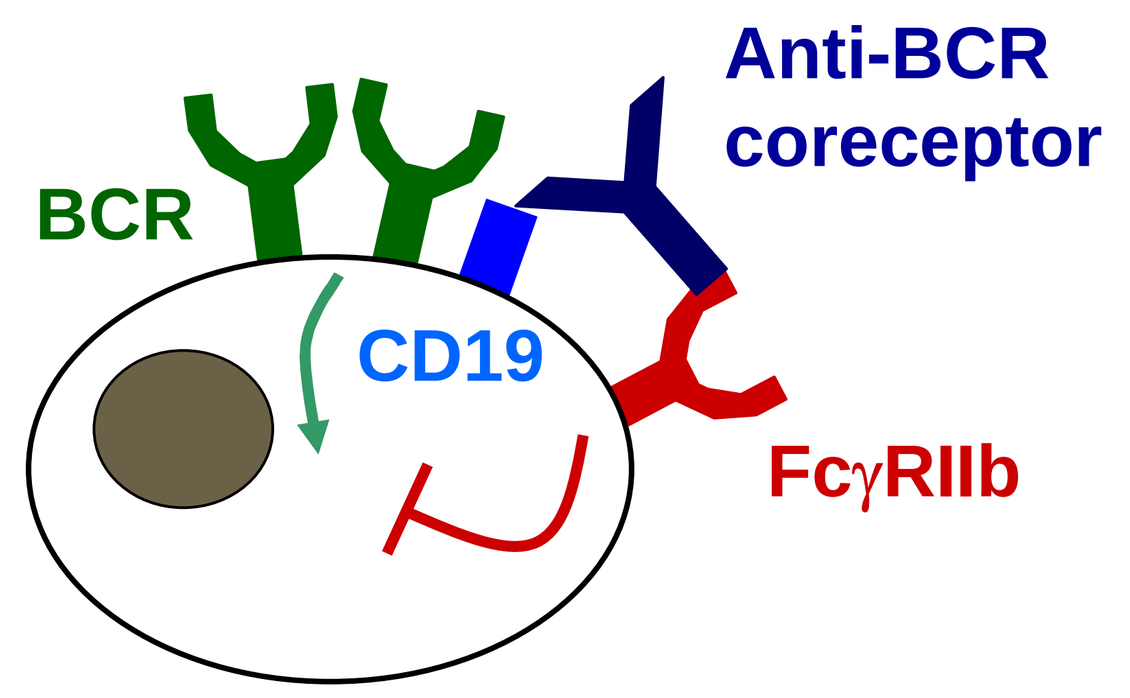
BCR
Anti-BCR coreceptor
CD19
FcγRIIb

Therapeutic strategy:

- Engineer a single antibody with a high-affinity Fab for BCR coreceptor complex (e.g., CD19) and a high-affinity Fc for inhibitory receptor FcγRIIb.
- Such an antibody may mimic the inhibitory effects of immune complexes on B cell function.

B Engineered Fc variants of human IgG1 have >2 logs improved affinity for FcγRIIb

1. FcγRIIb-selective variants identified from IgG1 Fc library screening

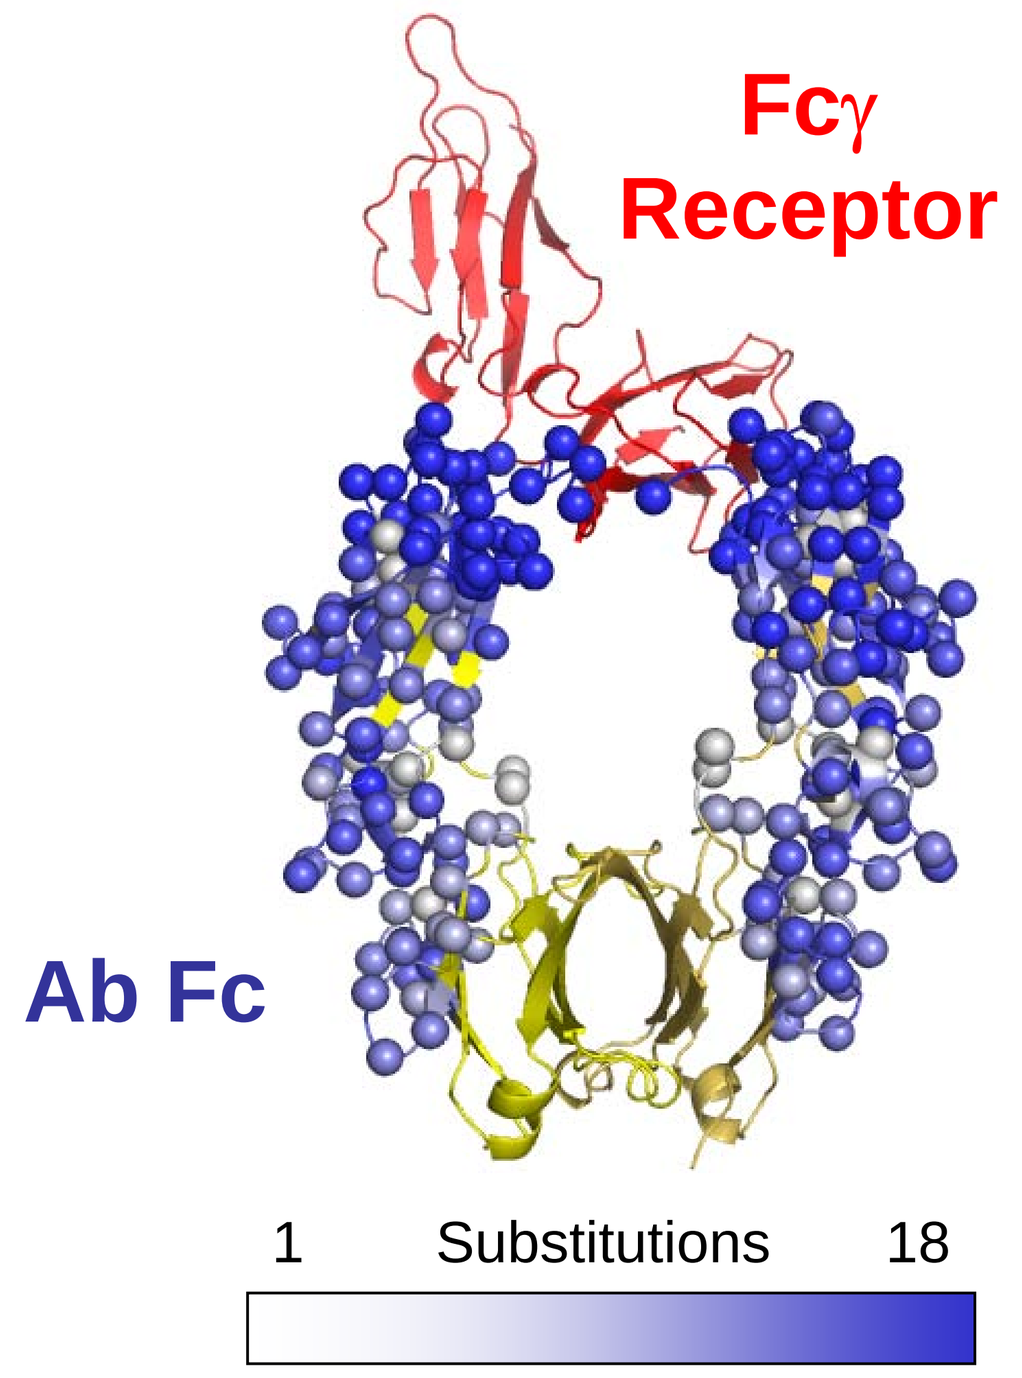
Fcγ Receptor
Ab Fc
1
Substitutions
18

2. Cell surface binding on FcγRIIb-transfected 293T cells

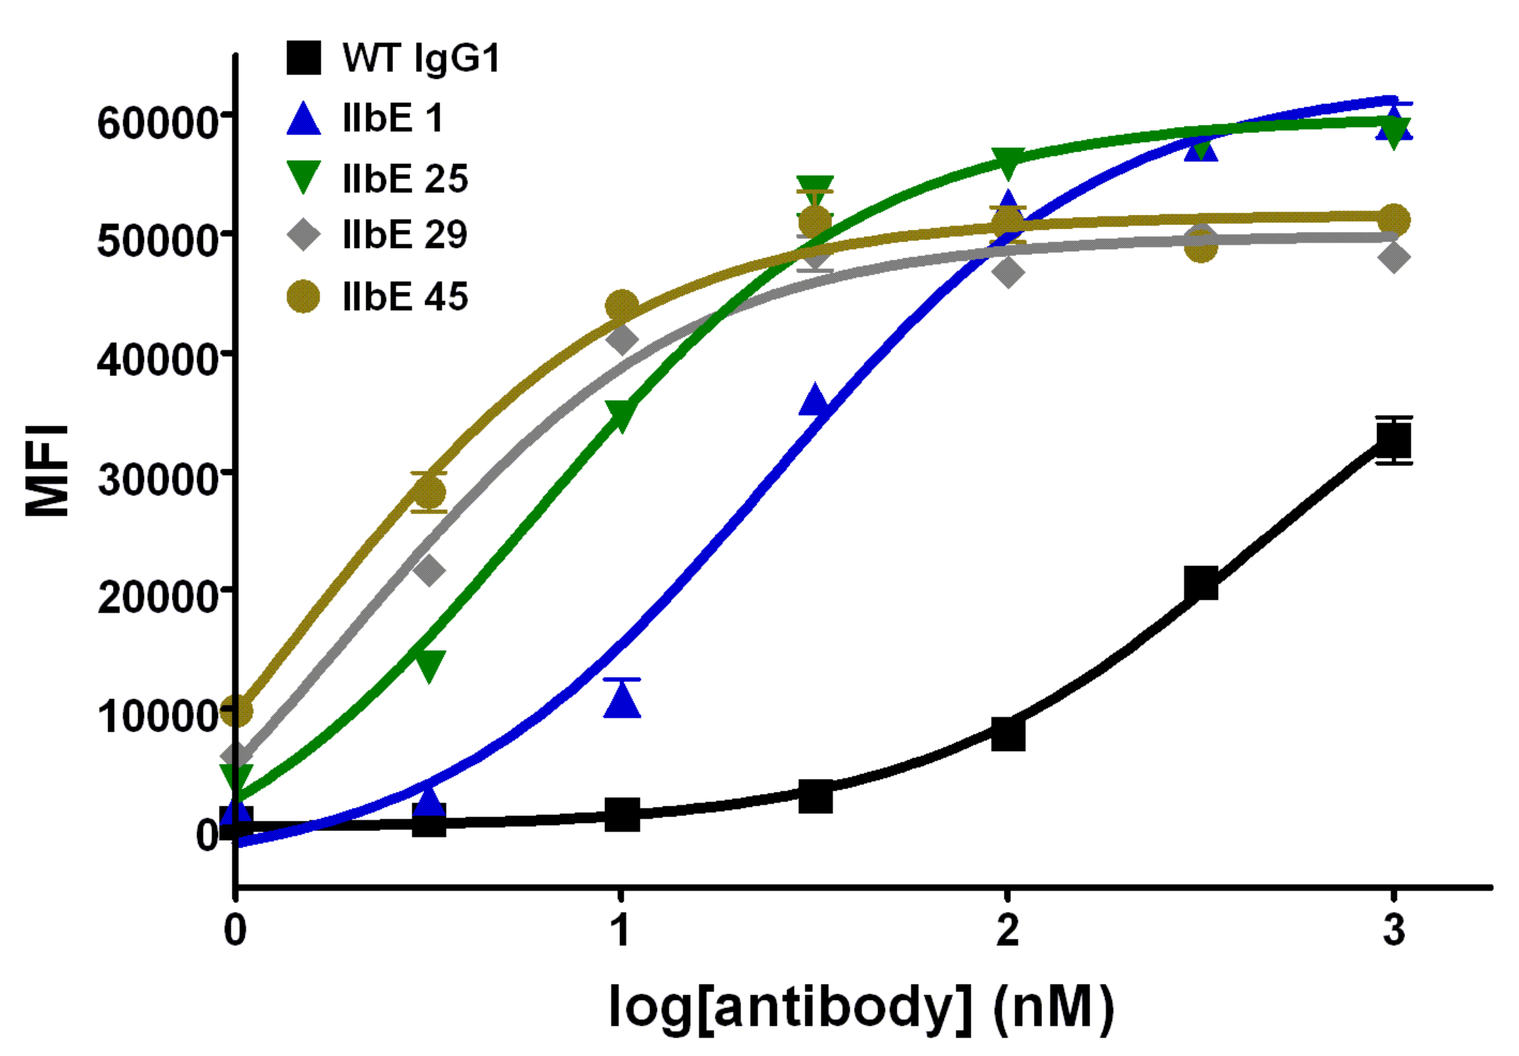
WT IgG1
IIbE 1
IIbE 25
IIbE 29
IIbE 45
MFI
log[antibody] (nM)

3. Anti-CD19 variants with enhanced FcγRIIb binding affinity ("IIbE")

Biacore Sensorgrams

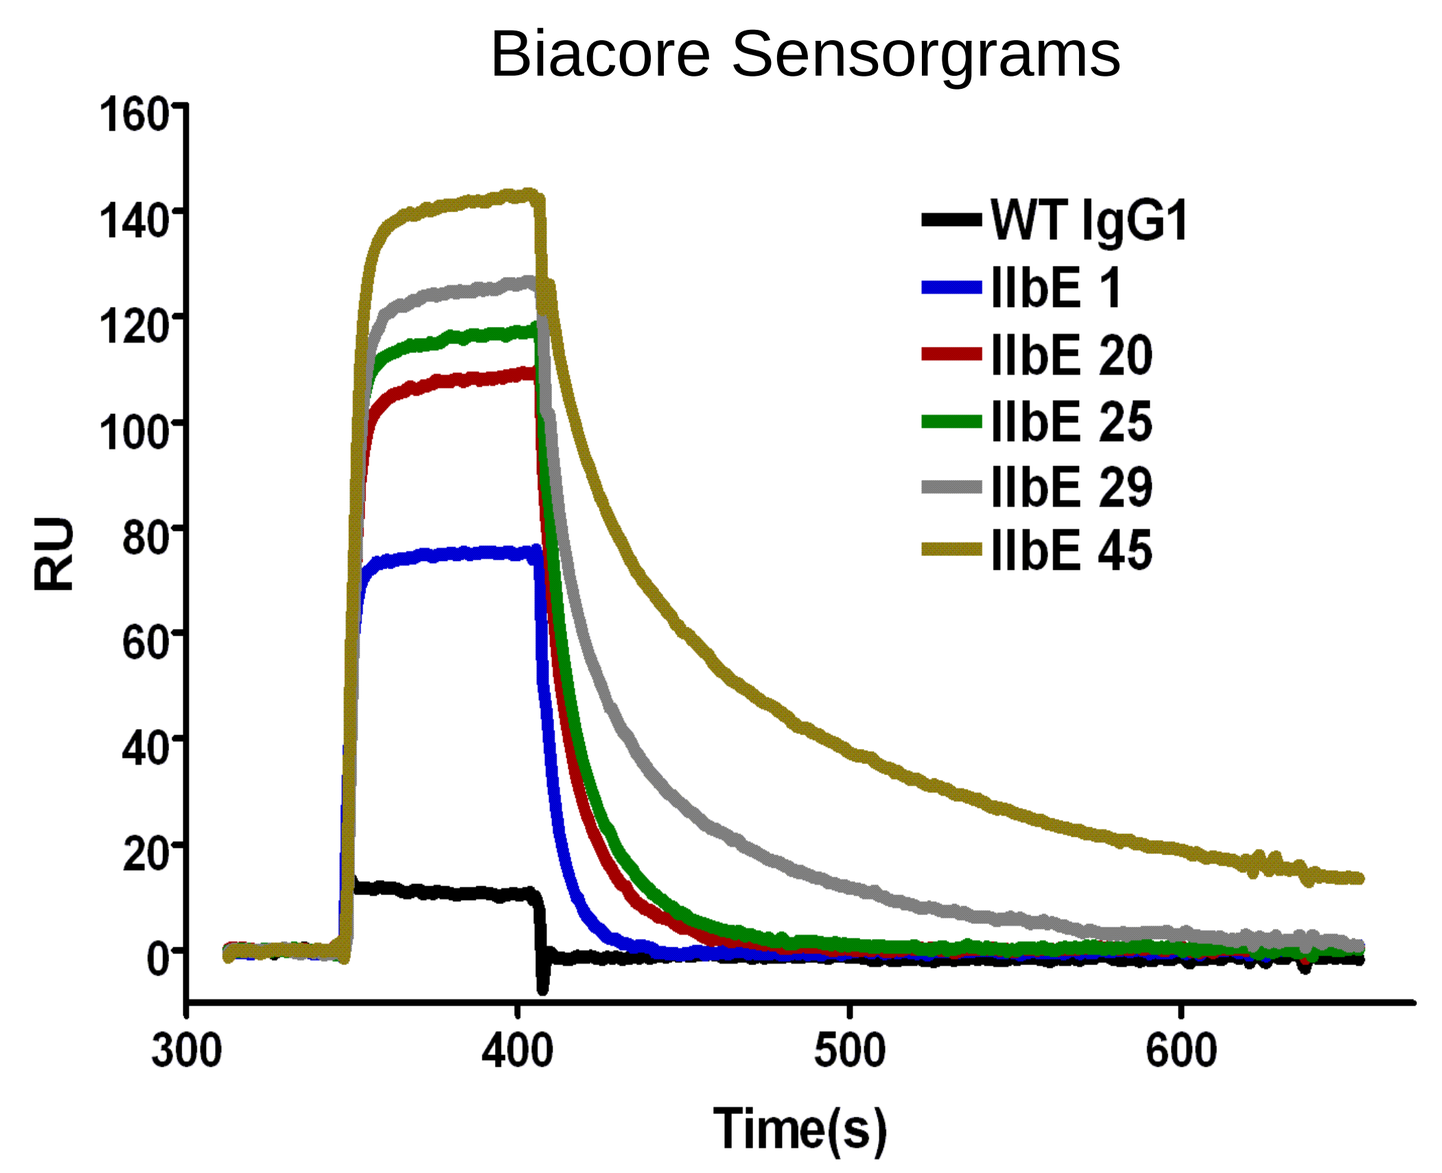
WT IgG1
IIbE 1
IIbE 20
IIbE 25
IIbE 29
IIbE 45
RU
Time(s)

4. Affinities of anti-CD19 IIbE variants for Fcγ Receptor IIb

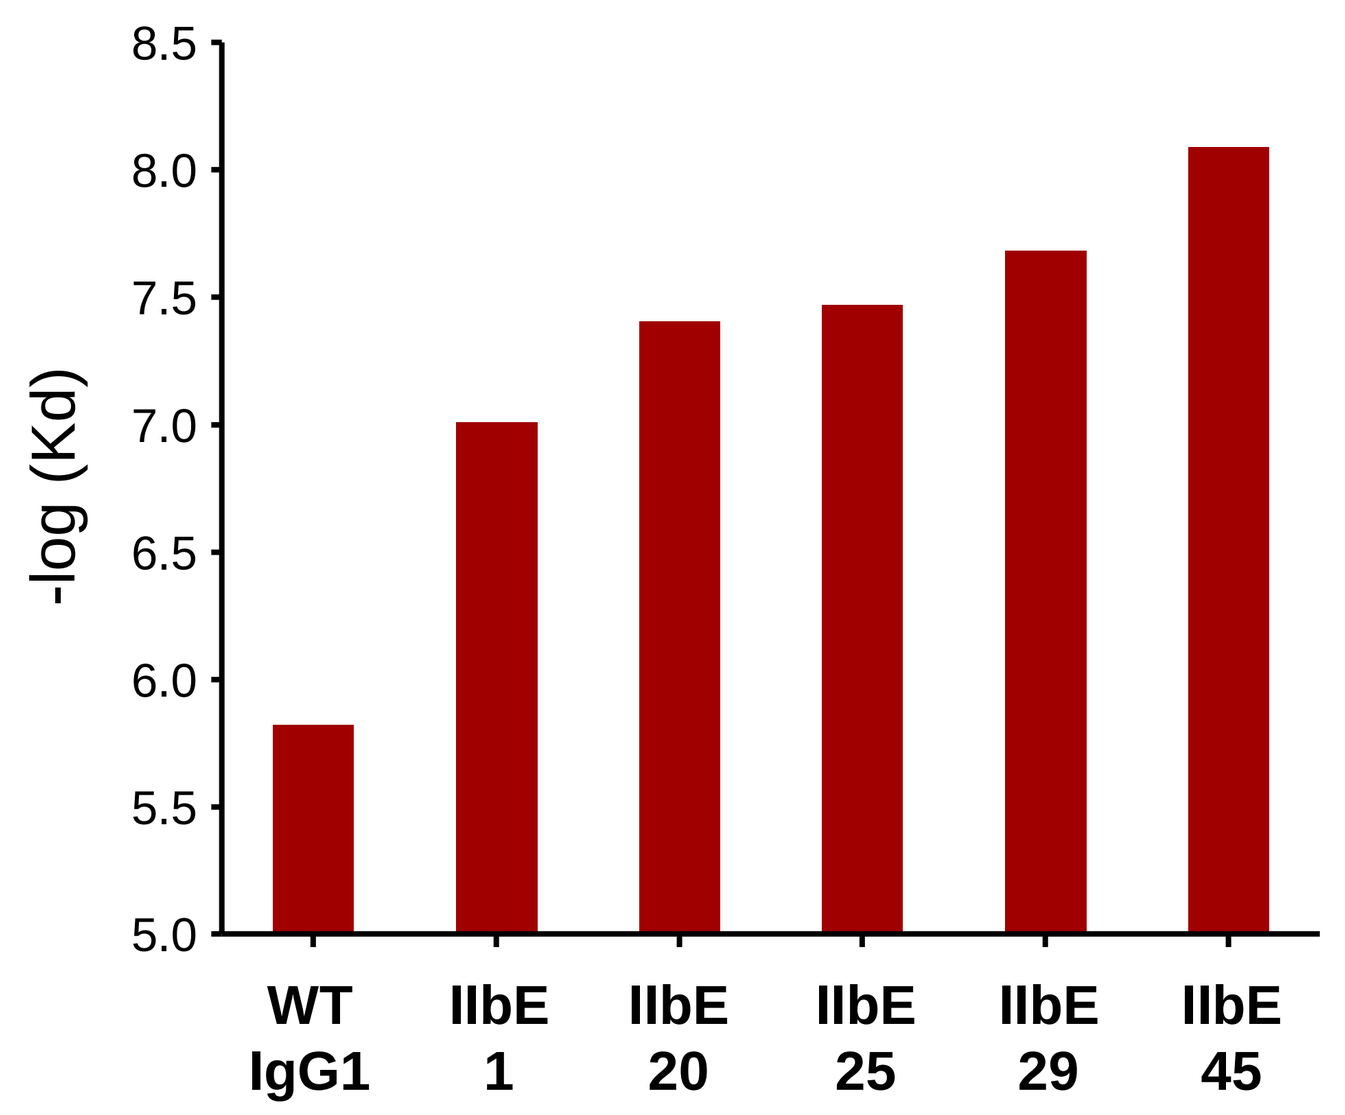
-log (Kd)
WT IgG1
IIbE 1
IIbE 20
IIbE 25
IIbE 29
IIbE 45

C Dual targeting of FcγRIIb and CD19 inhibits B cell calcium mobilization and viability

1. Inhibition of calcium flux in 1° B cells stimulated with anti-BCR by anti-CD19 antibodies with IIbE Fc domains

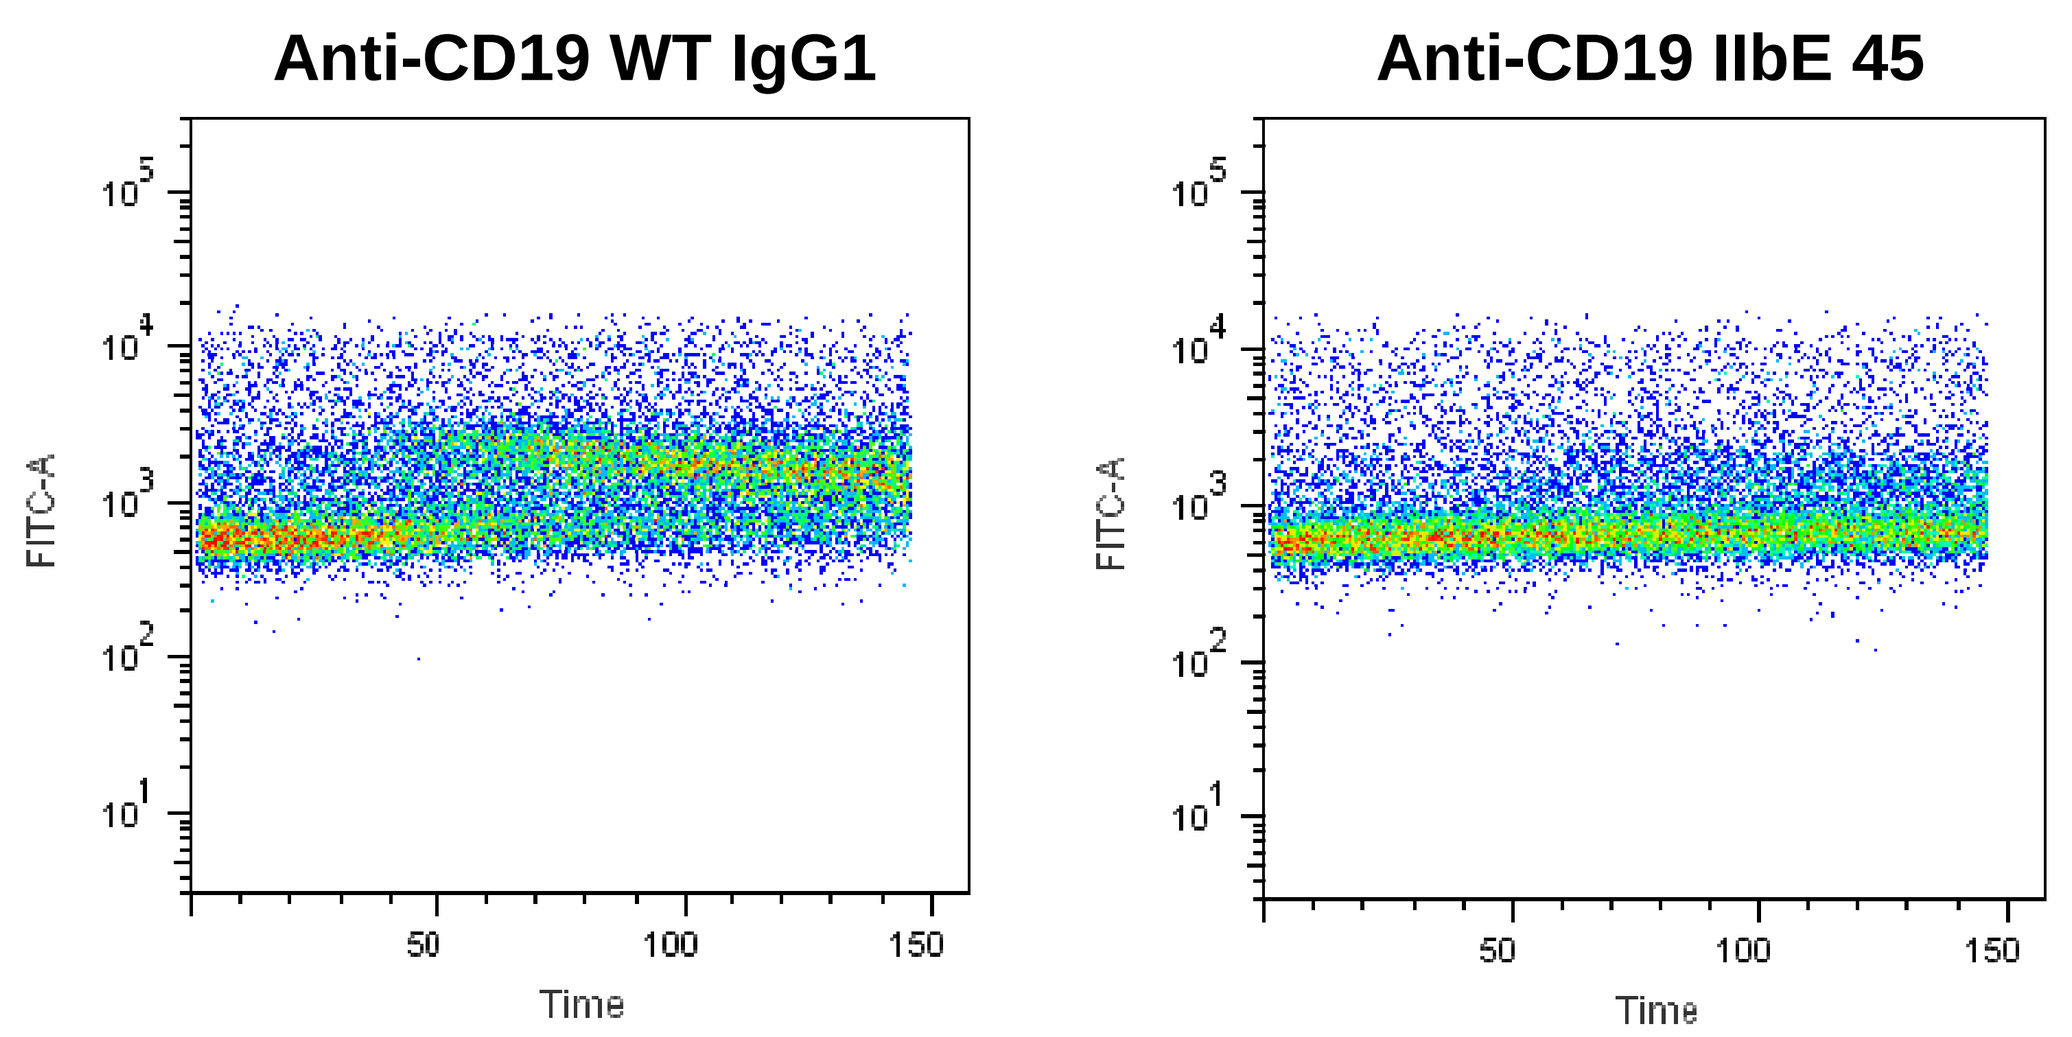
Anti-CD19 WT IgG1
Anti-CD19 IIbE 45
FITC-A
Time

Inhibition of calcium flux by anti-CD19 IIbE variants correlates with affinity for FcγRIIb

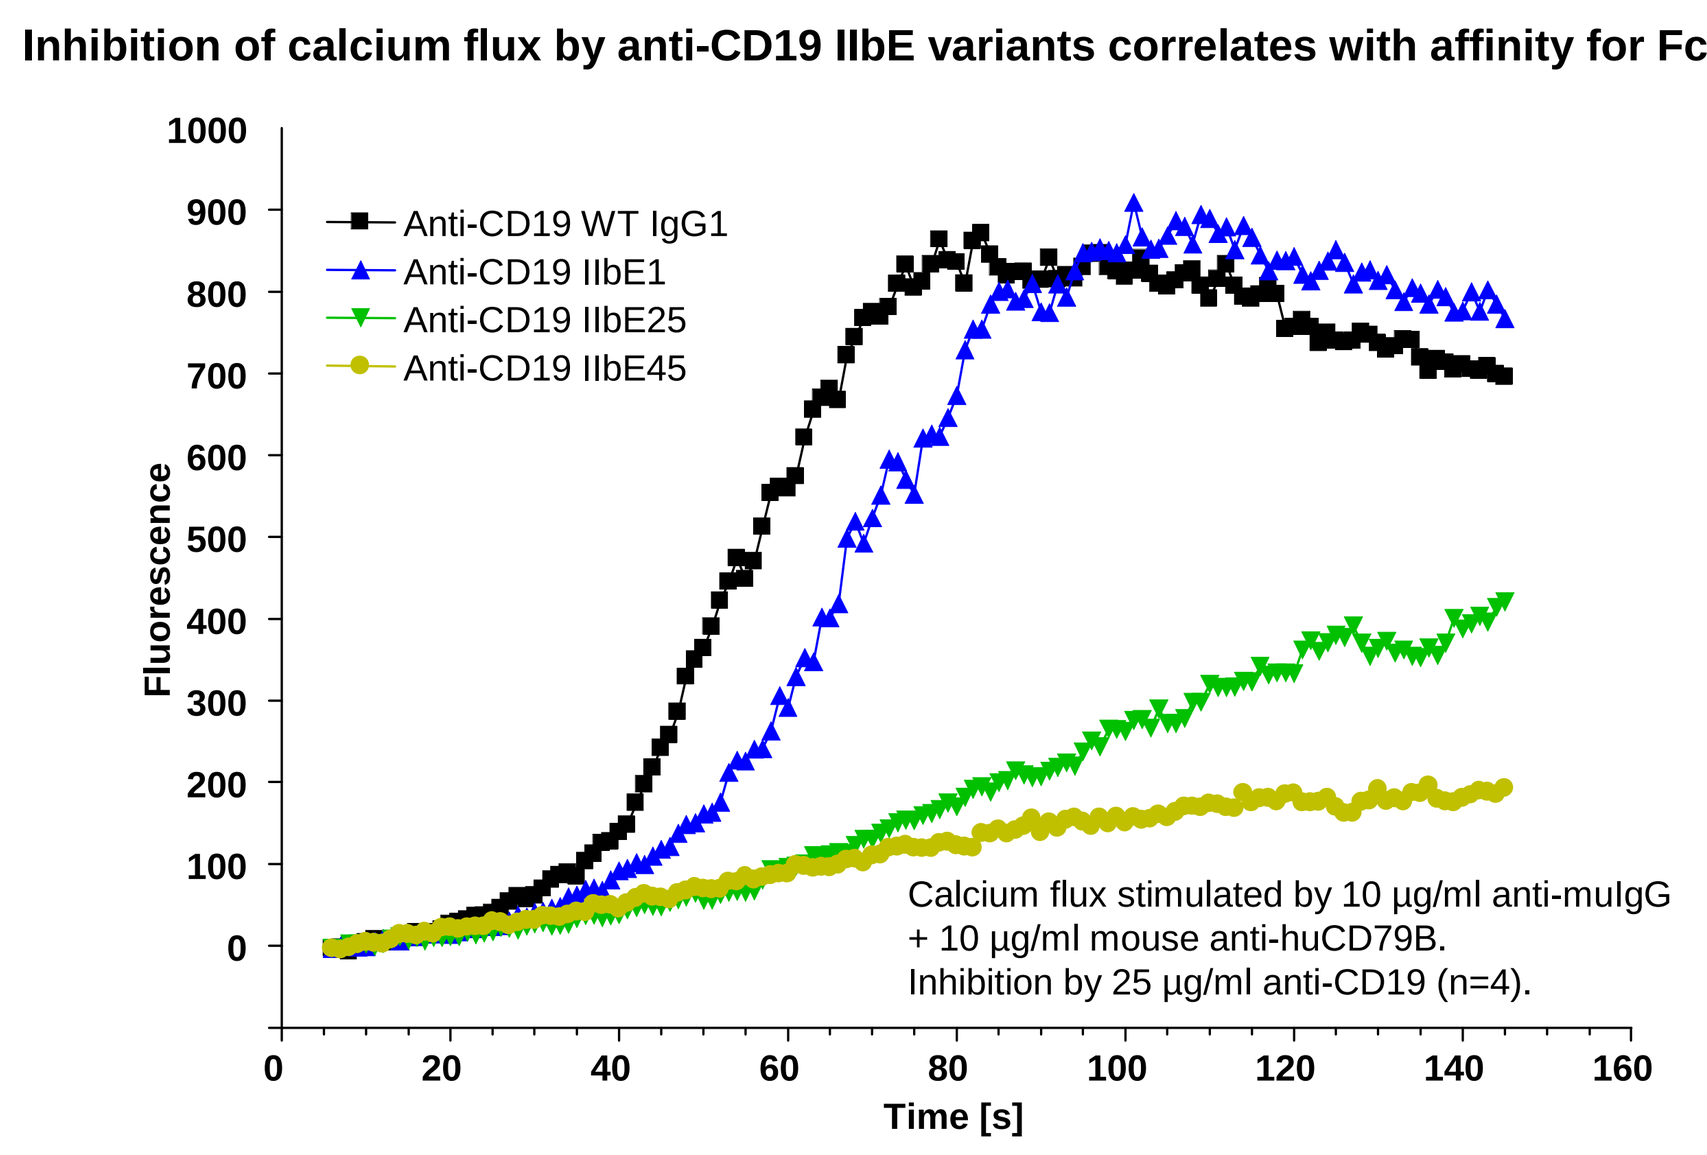
Anti-CD19 WT IgG1
Anti-CD19 IIbE1
Anti-CD19 IIbE25
Anti-CD19 IIbE45
Fluorescence
Time [s]
Calcium flux stimulated by 10 µg/ml anti-muIgG + 10 µg/ml mouse anti-huCD79B.
Inhibition by 25 µg/ml anti-CD19 (n=4).

Inhibition of calcium flux by anti-CD19 IIbE variant is dose-dependent

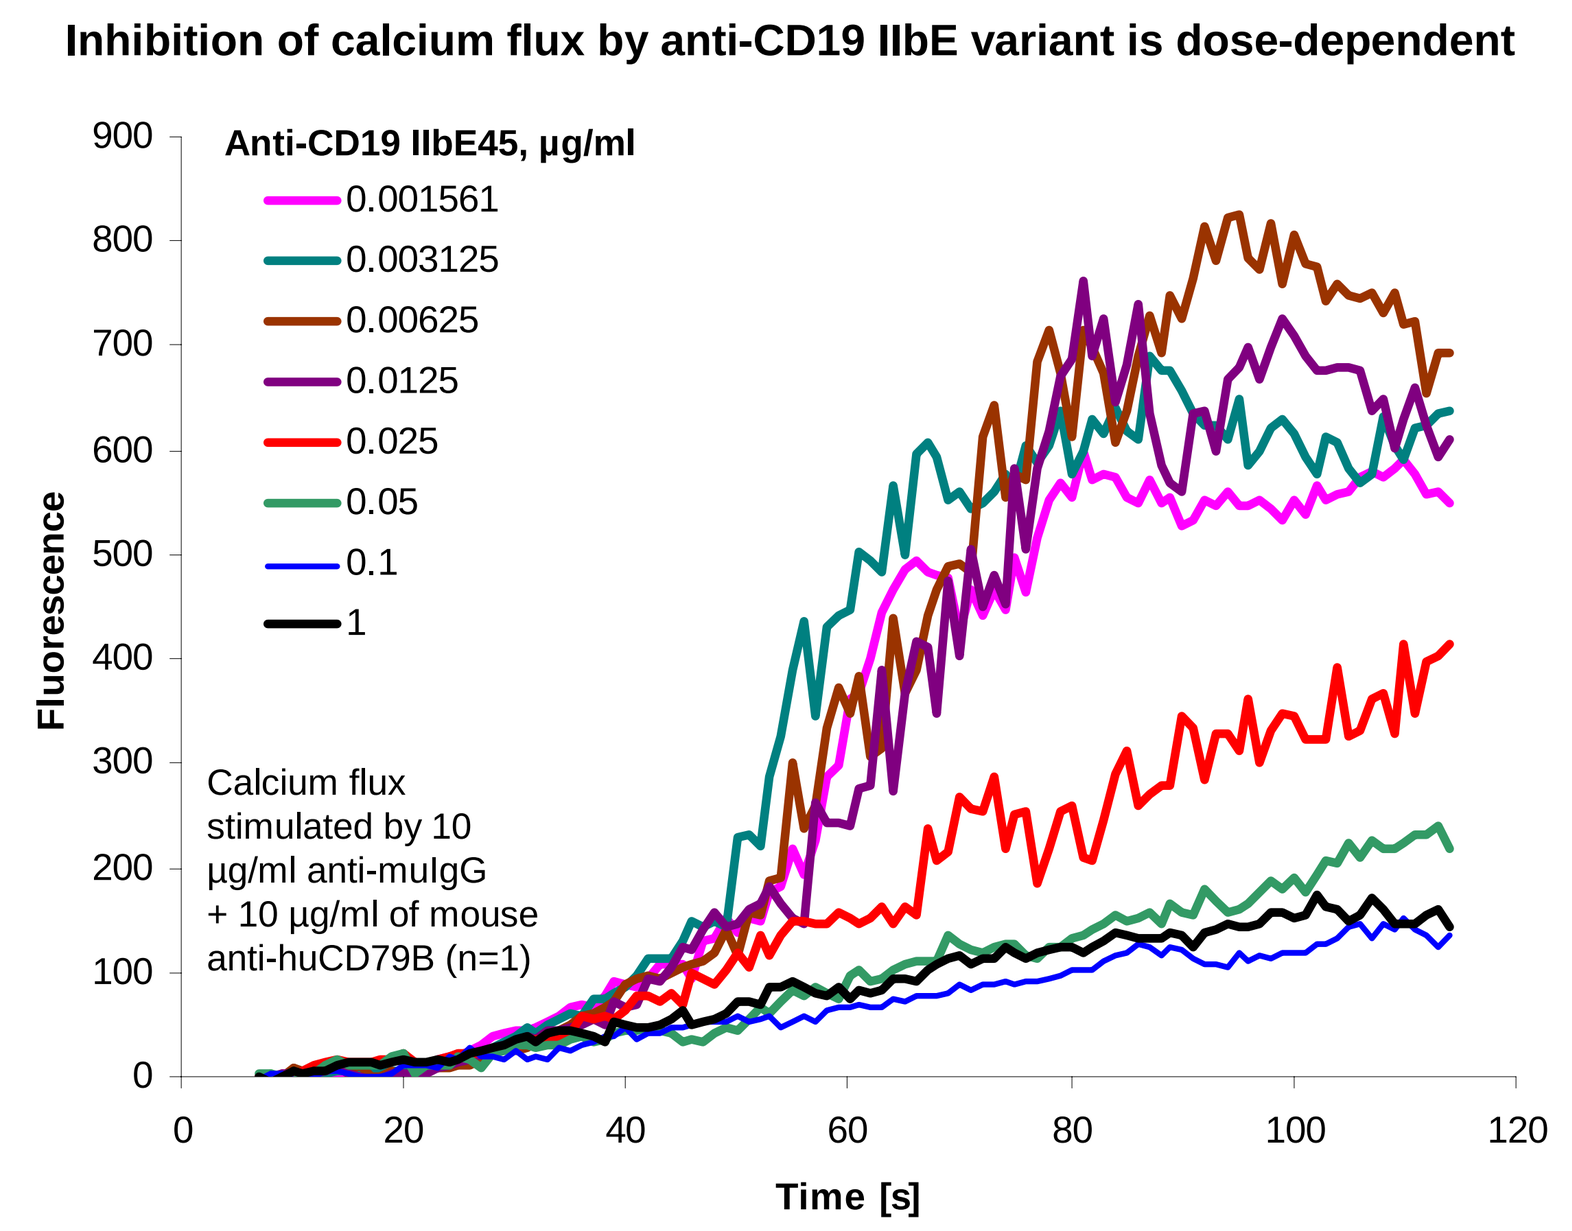
Anti-CD19 IIbE45, µg/ml
0.001561
0.003125
0.00625
0.0125
0.025
0.05
0.1
1
Fluorescence
Time [s]
Calcium flux stimulated by 10 µg/ml anti-muIgG + 10 µg/ml of mouse anti-huCD79B (n=1)

2. BCR-induced B cell viability is inhibited by dual targeting of CD19 and FcγRIIb using anti-CD19 IIbE

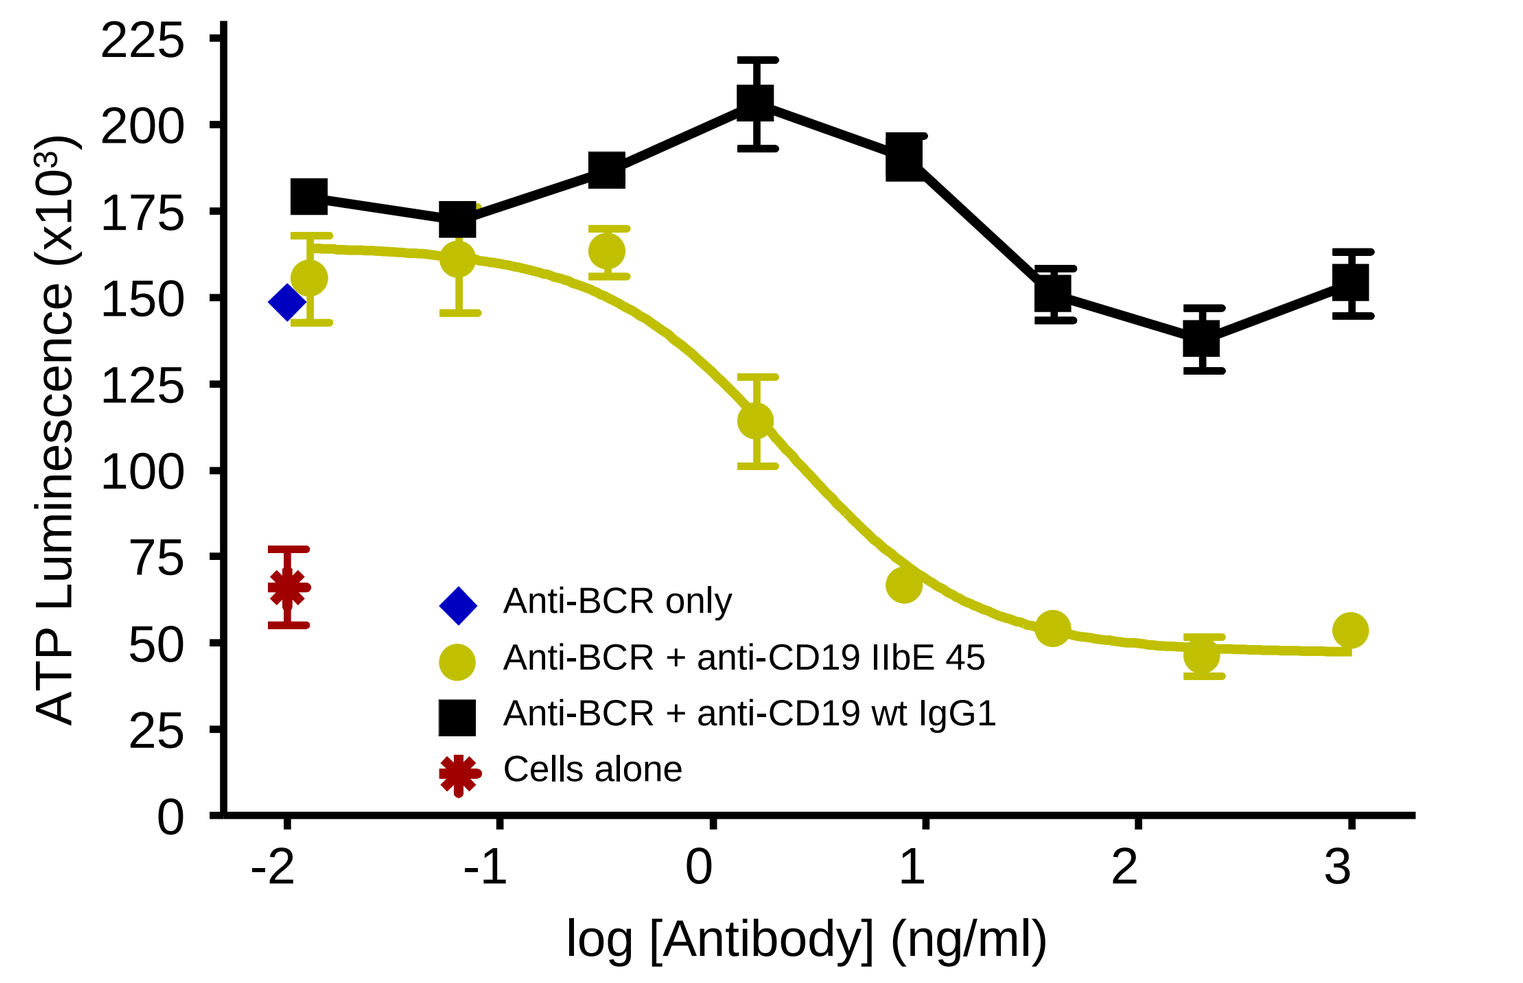
ATP Luminescence (x10³)
Anti-BCR only
Anti-BCR + anti-CD19 IIbE 45
Anti-BCR + anti-CD19 wt IgG1
Cells alone
log [Antibody] (ng/ml)

3. Inhibition of calcium mobilization requires FcγRIIb: Soluble FcγRI neutralizes Fc interaction

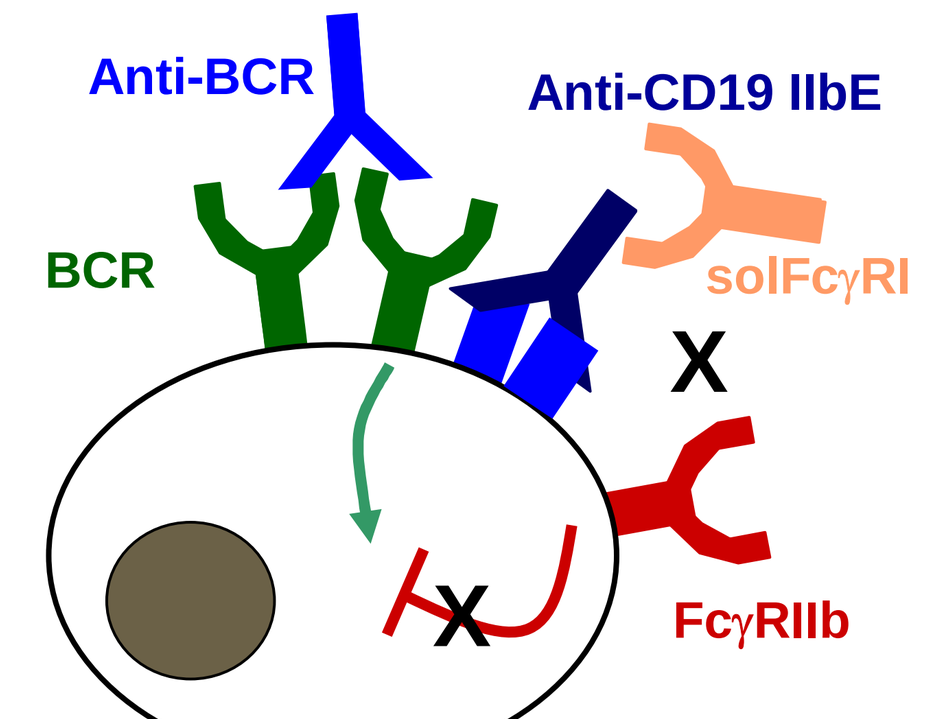
Anti-BCR
Anti-CD19 IIbE
BCR
solFcγRI
X
FcγRIIb

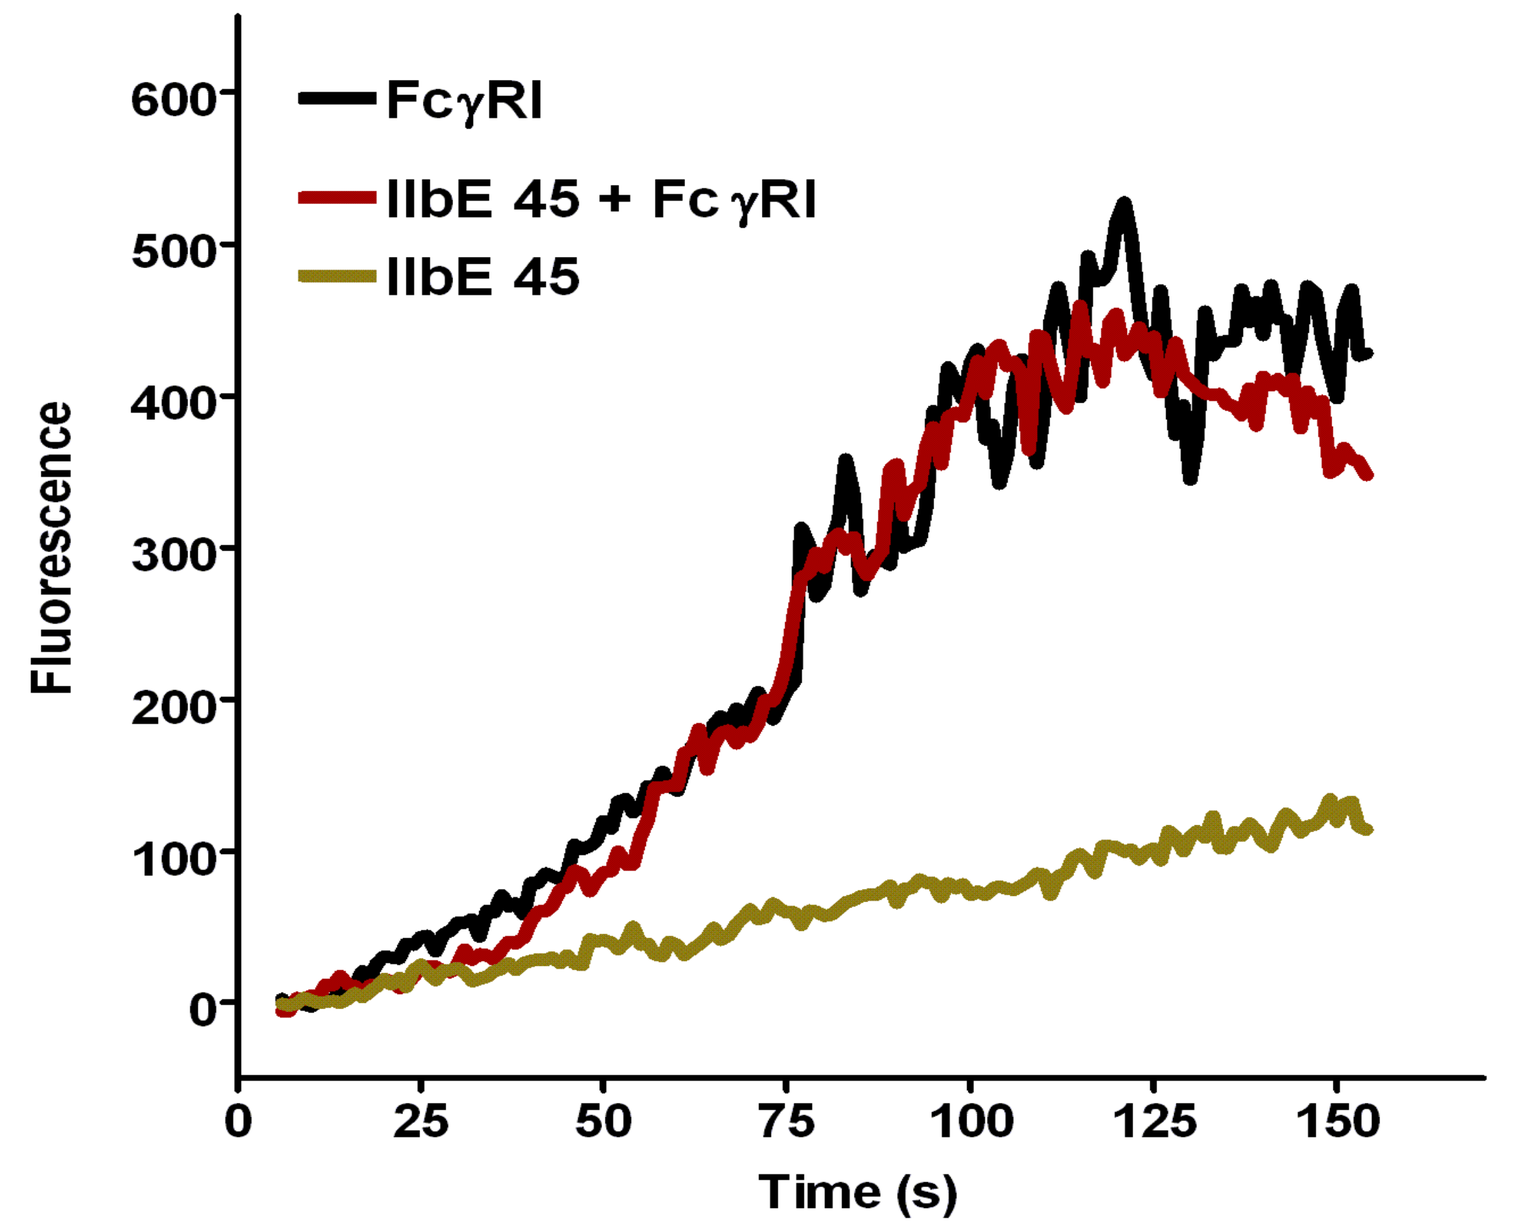
FcγRI
IIbE 45 + Fc γRI
IIbE 45
Fluorescence
Time (s)

D IIbE variants reduce immune-mediated effector functions

1. B cell depletion by ADCC (NK cell mediated): Anti-CD19 IIbE does not stimulate

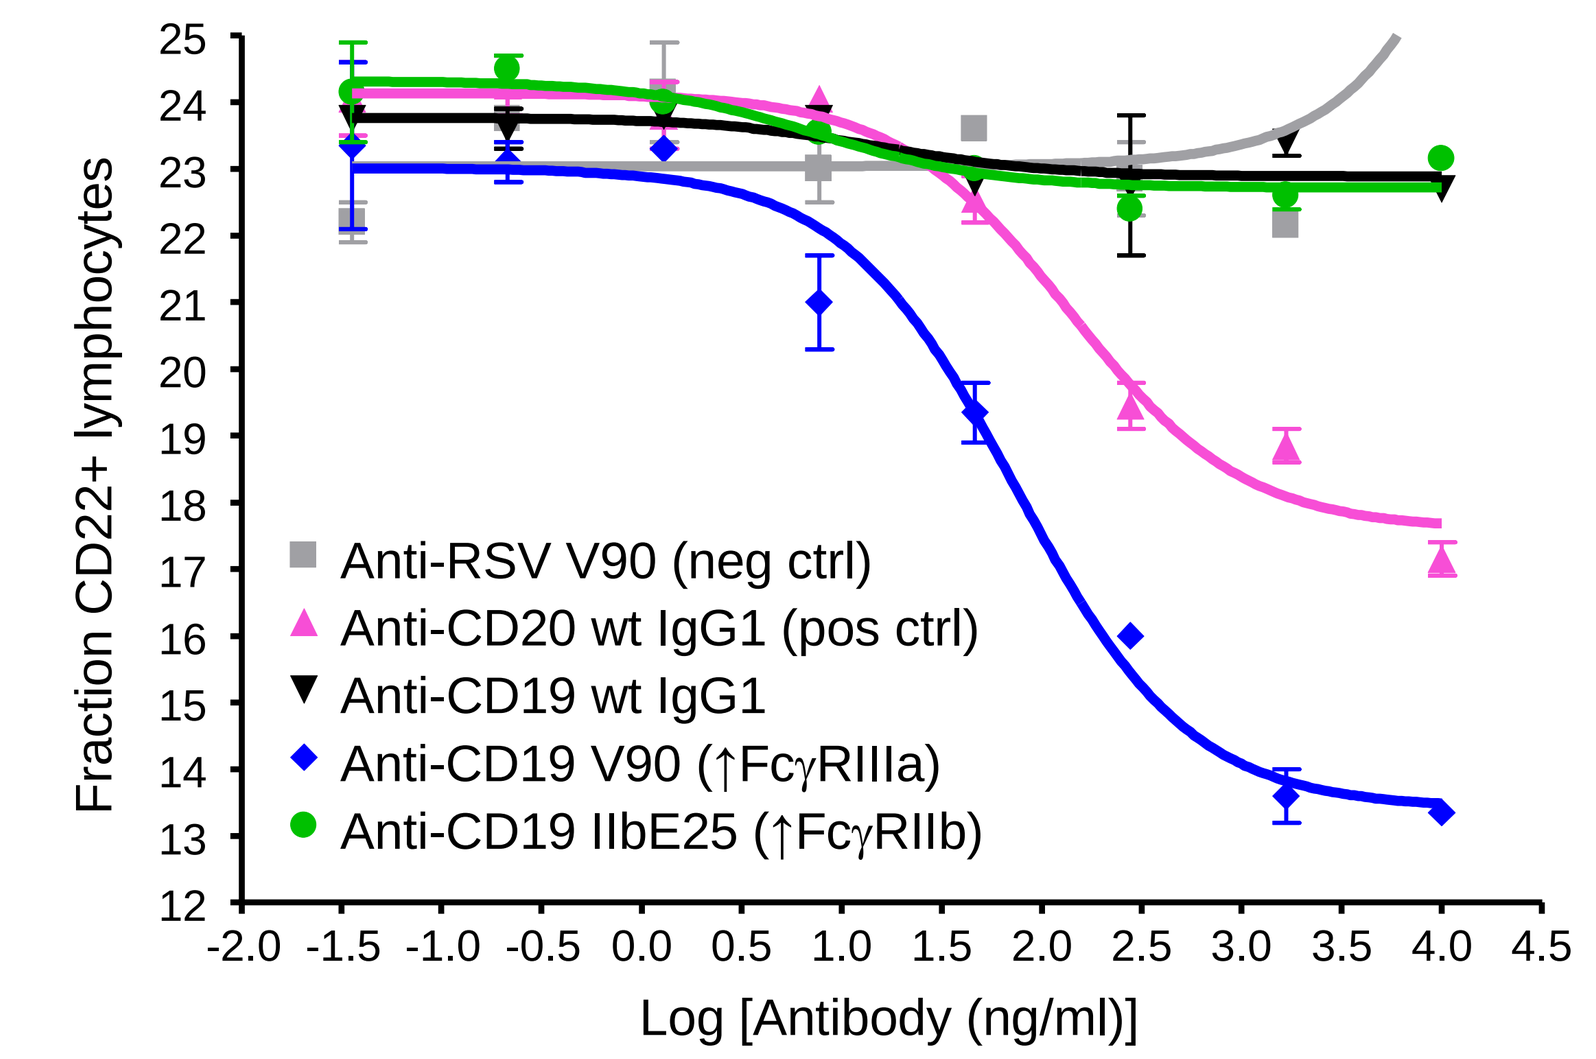
Fraction CD22+ lymphocytes
Anti-RSV V90 (neg ctrl)
Anti-CD20 wt IgG1 (pos ctrl)
Anti-CD19 wt IgG1
Anti-CD19 V90 (↑FcγRIIIa)
Anti-CD19 IIbE25 (↑FcγRIIb)
Log [Antibody (ng/ml)]

2. B cell depletion by ADCP (macrophage-mediated): Anti-CD19 IIbE does not stimulate

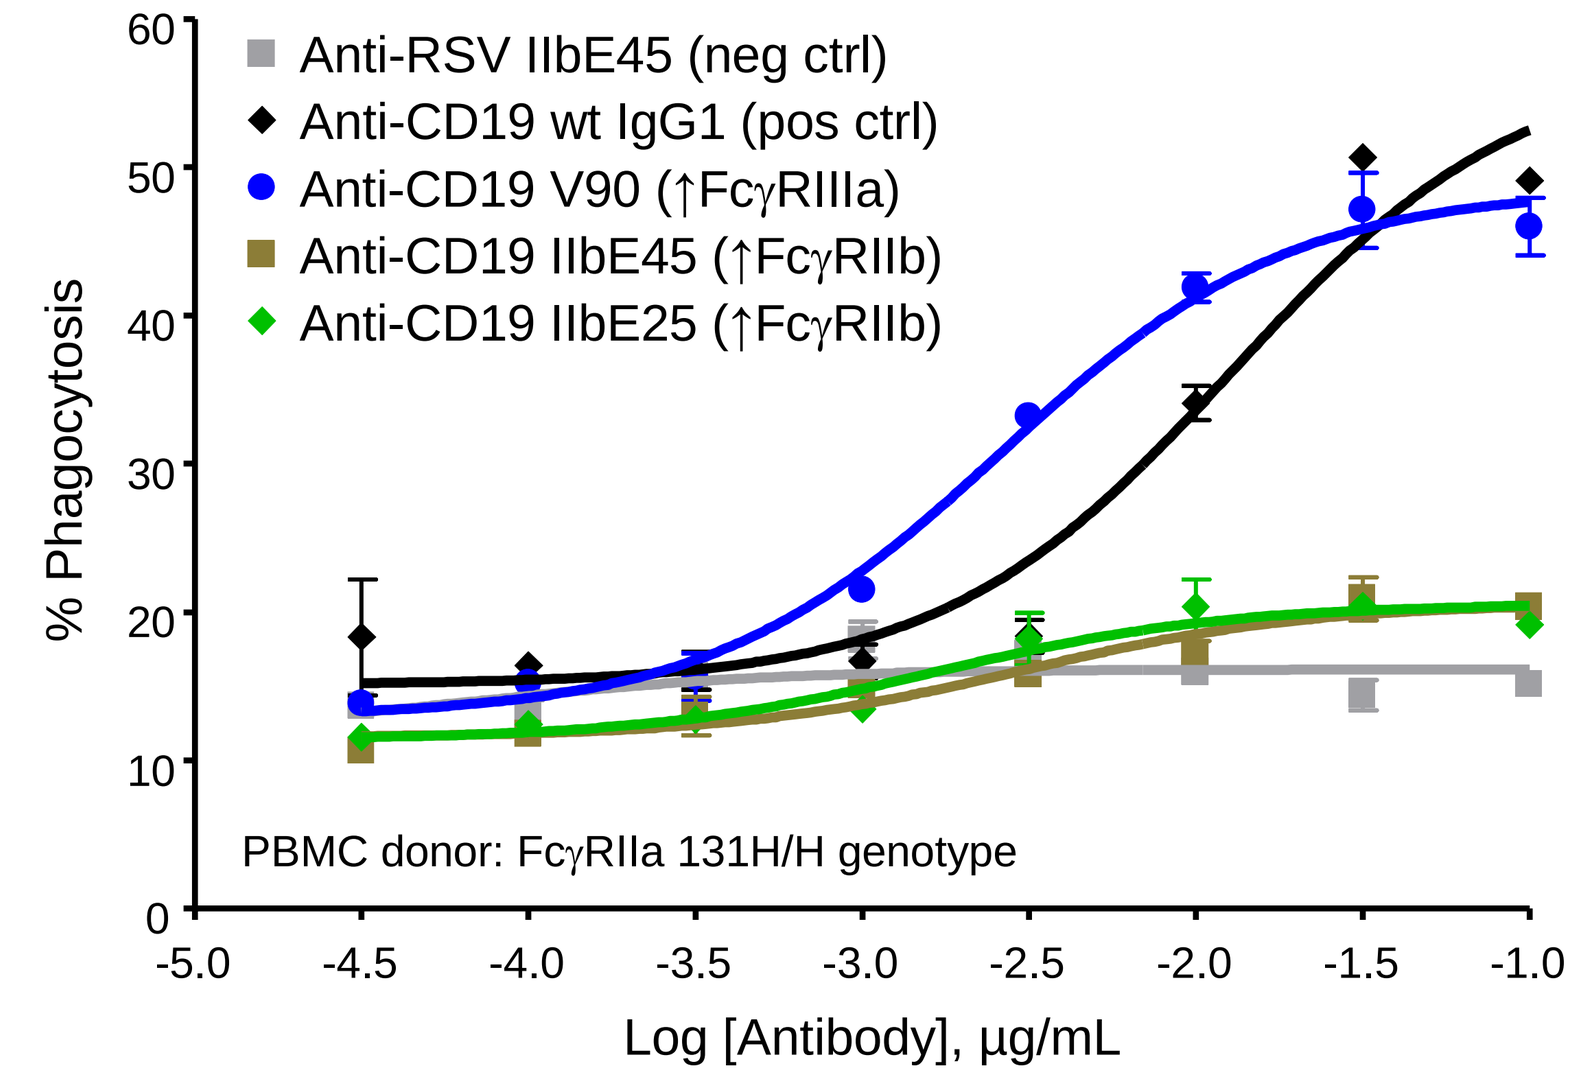
% Phagocytosis
Anti-RSV IIbE45 (neg ctrl)
Anti-CD19 wt IgG1 (pos ctrl)
Anti-CD19 V90 (↑FcγRIIIa)
Anti-CD19 IIbE45 (↑FcγRIIb)
Anti-CD19 IIbE25 (↑FcγRIIb)
PBMC donor: FcγRIIa 131H/H genotype
Log [Antibody], µg/mL

3. Anti-CD19 IIbE does not possess CDC activity

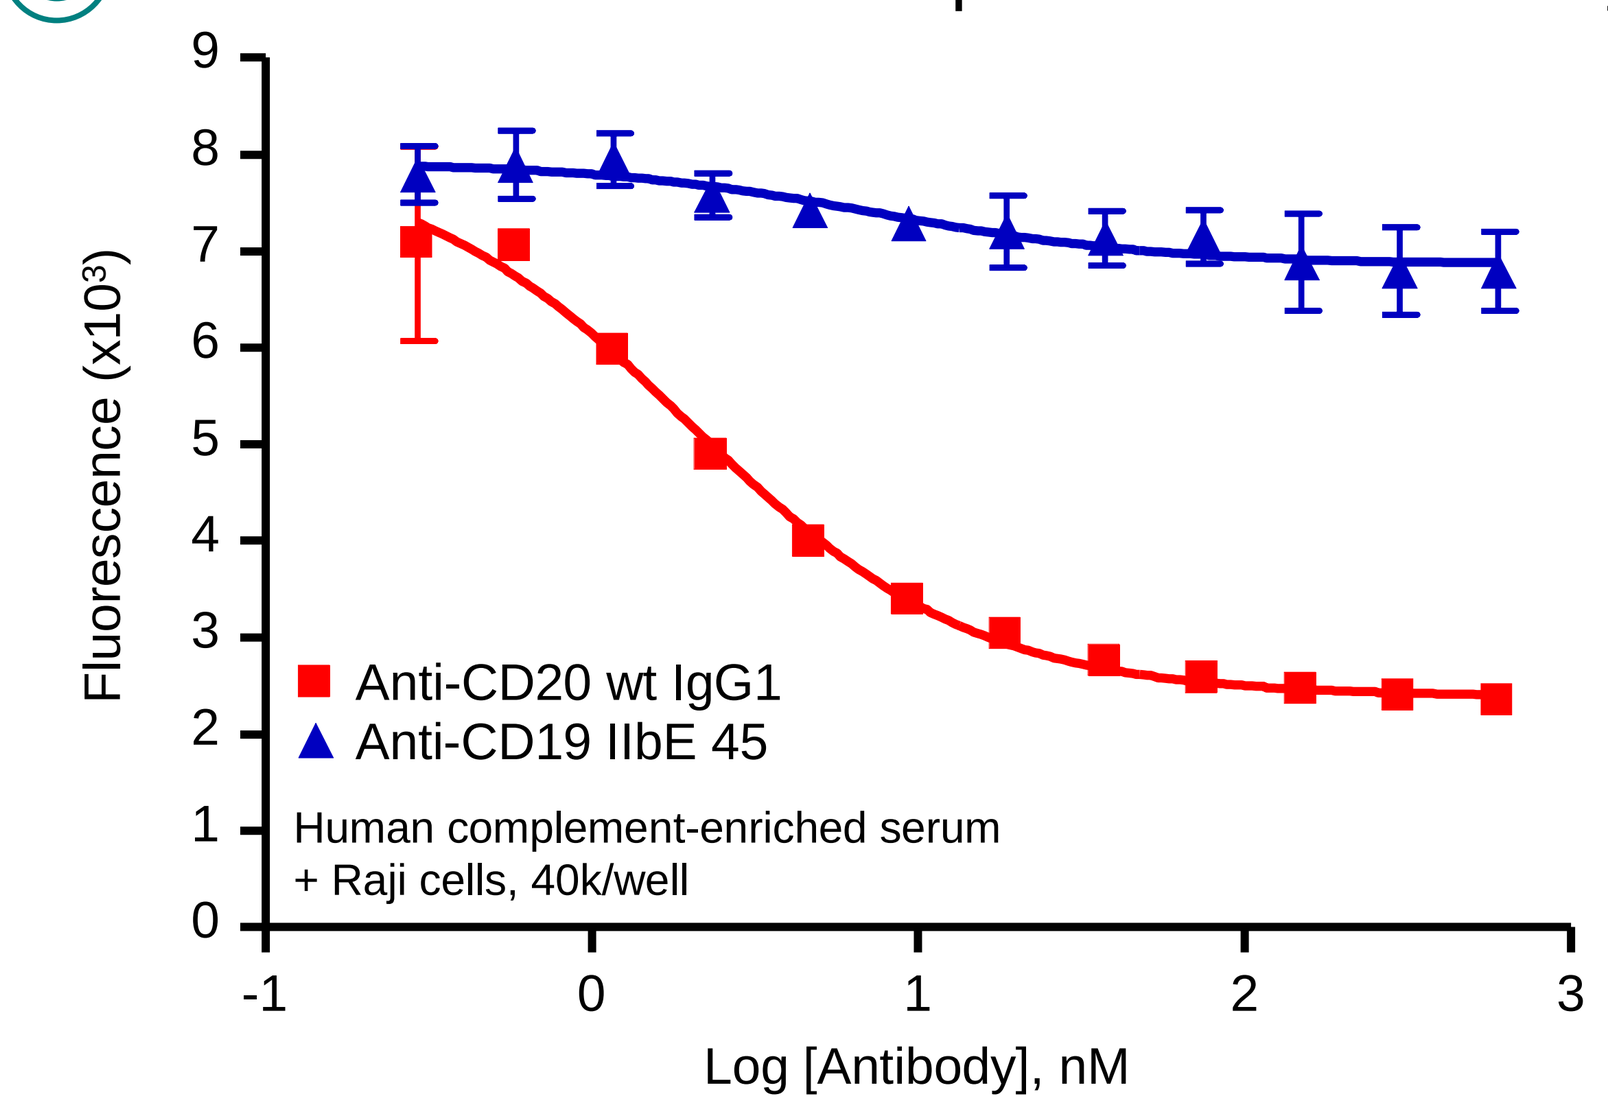
Fluorescence (x10³)
Anti-CD20 wt IgG1
Anti-CD19 IIbE 45
Human complement-enriched serum
+ Raji cells, 40k/well
Log [Antibody], nM

Summary

- We have engineered FcγRIIb-enhanced ("IIbE") humanized anti-BCR coreceptor antibodies with high affinity for both FcγRIIb (8 nM) and CD19 (0.5 nM):
 - Anti-CD19 IIbE antibodies inhibited B cell activation responses including anti-BCR-induced calcium mobilization and in vitro viability.
 - Anti-CD19 IIbE antibodies showed reduced immune-mediated effector functions, including NK cell-mediated ADCC and macrophage-mediated ADCP.
- The required coengagement of FcγRIIb and CD19 by a single antibody increases B cell specificity and potentially reduces off-target effects.
- The IIbE mechanism represents a potential novel therapeutic strategy to downmodulate multiple B cell functions in autoimmune disease.

© 2007 Xencor, Inc. Monrovia, California 91016 USA; *david.szymkowski@xencor.com