XmAb™2513, an Fc-engineered humanized anti-CD30 monoclonal antibody, has potent *in vitro* and *in vivo* activities, and has the potential for treating hematologic malignancies

Chris E. Lawrence[1], Philip W. Hammond [1]*, Jonathan Zalevsky [1]*, Holly Horton [1]*, Seung Chu [1]*, Sher Karki [1]*, John R. Desjarlais [1]*, David F. Carmichael [1]*.*

[1] Introduced by Dr. Eugene Zhukovsky, * Xencor Inc., Monrovia, CA 91016, USA, Contact: *clawrence@xencor.com*

Abstract

XmAb[TM]2513 is a novel humanized monoclonal antibody (mAb) that binds to the human cell surface antigen CD30 and demonstrates anti-proliferative activity against CD30-positive (CD30+) cell lines. XmAb2513 also has an engineered Fc region to enhance cell killing activity via recruitment of effector cells through increased binding affinity to Fcγ receptors (FcγRs). Consequently, XmAb2513 exhibits superior antibody-dependent cell-mediated cytotoxicity (ADCC) and antibody-dependent cell-mediated phagocytosis (ADCP), when compared to a native IgG1 (unengineered) version of the antibody.

To evaluate the potential clinical activity of XmAb2513 in CD30+ diseases such as Hodgkin Lymphoma (HL) and Anaplastic Large Cell Lymphoma (ALCL), XmAb2513 was tested in murine subcutaneous xenograft models of HL using the CD30+ L540 cell line. In the ICR-SCID mouse strain, intraperitoneal (ip) administration of XmAb2513 at 3 mg/kg every 4 days for 10 doses (q4d x10), gave a statistically significant reduction in tumor growth and enhanced survival relative to the control. At doses of 10 and 30 mg/kg XmAb2513 (ip, q4d x10) tumor growth was not only slowed, but elimination of established tumors was observed in 3/9 and 5/9 animals respectively. The treatment was well-tolerated.

Preclinical studies were conducted to evaluate the safety and pharmacokinetics of XmAb2513 in large animals. In vitro studies demonstrated that the cynomolgus monkey was the appropriate species for study. Binding affinities of XmAb2513 to both human and cynomolgus monkey CD30 and FcγRs were evaluated by Biacore methods and were found to be similar. Additionally, fluorescein-XmAb2513 gave similar staining patterns in immunohistochemistry cross-reactivity studies with normal human and cynomolgus monkey tissue panels. As evidenced by in-life observations, single (0, 1 and 100 mg/kg XmAb2513, intravenous [iv] infusion) and repeat dose (0, 10, 30 and 100 mg/kg XmAb2513, q5d x6, iv infusion) treatment with XmAb2513 was well-tolerated. Serum cytokines showed no trend that was indicative of an XmAb2513-related effect following single dose administration.

The pharmacokinetics of XmAb2513 was also determined after either single or repeat dose administration. In the repeat dose study (0, 3, 10 and 30 mg/kg XmAb2513, q5d x6, iv infusion) exposure increased in a dose proportional manner, and terminal half-life ($t_{½}$) ranged from 12-17 days. In the single dose study (0, 1 and 100 mg/kg XmAb2513, iv infusion) exposure was also proportional to dose. The exposure and $t_{½}$ data support at least an every other week dosing interval in the clinic.

These preclinical data provide a rationale for the clinical testing of XmAb2513 in patients with hematologic malignancies that express CD30, specifically HL and ALCL, and support the safety of repeat administration of XmAb2513 in humans.

XmAb2513 (anti-CD30 mAb)

- Humanized version of the murine mAb AC10 (anti-CD30)
 - cluster C epitope
- XmAb Fc region
 - engineered for high affinity Fcγ receptor binding
- Antiproliferative against
 - L540 cell line (Hodgkin Lymphoma)
 - Karpas 299 cell line (Anaplastic Large Cell Lymphoma)
- Blocks CD30 ligand (CD30L) binding
- Has improved effector functions
 - Cytotoxicity (Antibody dependent cell-mediated cytotoxicity, ADCC)
 - Phagocytosis (Antibody dependent cell-mediated phagocytosis, ADCP)
- Does not exhibit complement dependent cytotoxicity (CDC)

XmAb2513 has comparable binding to Human and Cyno solCD30-Fc and FcγRs

Soluble CD30-Fc

	Human	Cynomolgus Monkey
	K_D (nM)	
sCD30-Fc	0.47	0.29

Fcγ Receptors

	Human	Cynomolgus Monkey
	K_D (nM)	
FcγRI	0.92	2.6
FcγRIIA	147	139
FcγRIIB	120	75
FcγRIIIA	9.1	4.8
FcRn	14.2	28.8

- The binding affinities of XmAb2513 to both human and cynomolgus monkey solCD30-Fc, and FcγR and FcRn receptors were very similar
- The rank order of affinities among the FcγR and FcRn receptors was identical between the two species
- The similarity of binding characteristics between human and cynomolgus monkey is consistent with the latter being the appropriate species for further evaluating the pharmacological and toxicological properties of XmAb2513

XmAb2513 increased survival in L540 Xenografts

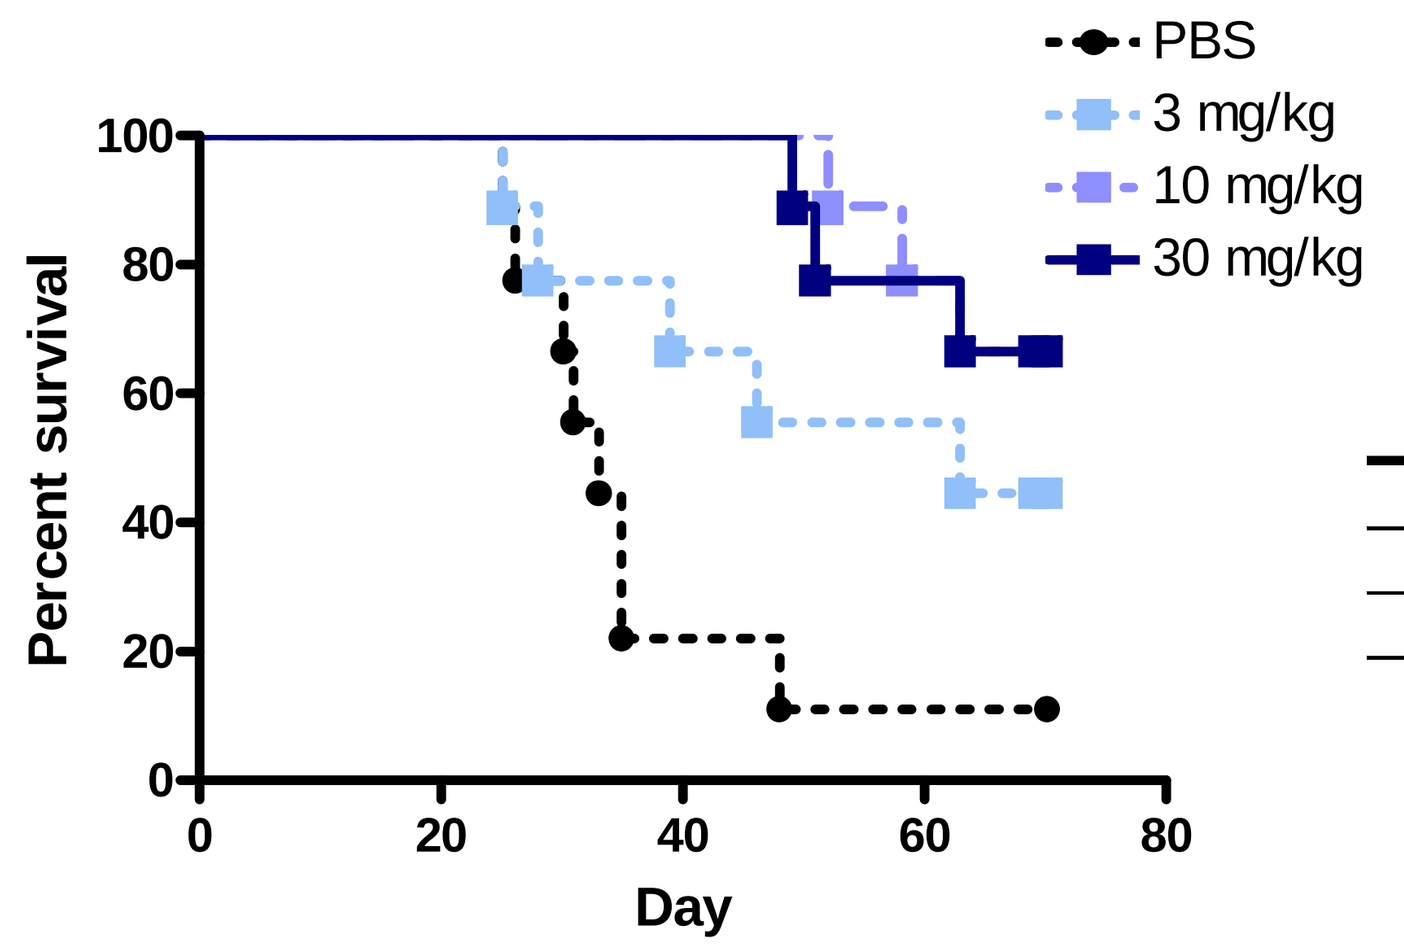
PBS
3 mg/kg
10 mg/kg
30 mg/kg
Percent survival
Day

Treatment (XmAb[TM]2513 mg/kg)	Mean Day to Reach Tumor Volume of 1000 mm^3 ±SD[1]	T-C in Days[2] (% Change)
0	30.0 ± 16.1	-
3	46.6 ± 20.3	16.6 (55%)
10	60.0 ± 10.2	30.0 (100%)
30	61.2 ± 14.3	31.2 (104%)

[1] Mean of results (n=9) from individual data
[2] Tumor growth delay (T-C) is calculated by subtracting the mean day to reach 1000mm³ (or to the day of the last tumor measurement) of the control group (C) from the mean day reached by the treated group (T). The % change is as compared to the control group.

XmAb2513 caused tumor regression in L540 Xenografts

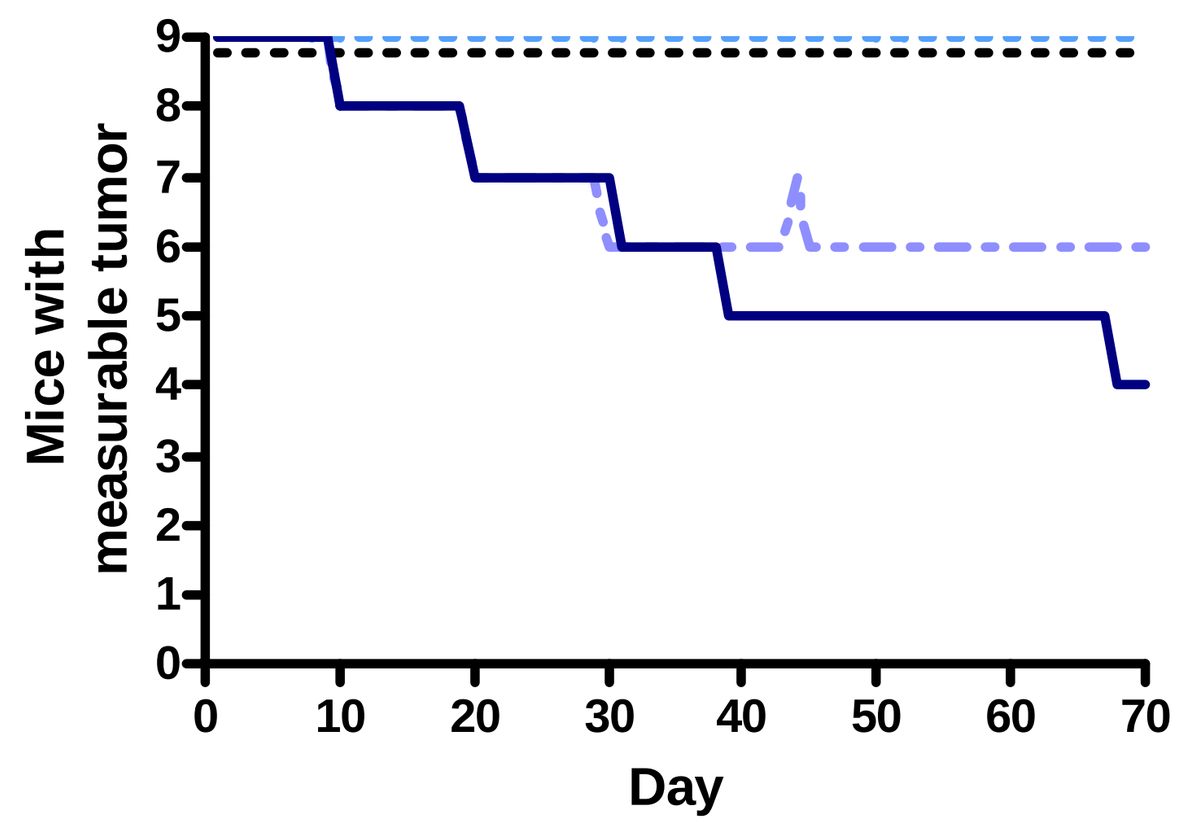
Mice with measurable tumor
Day

	Regressions (of 9 mice)
PBS	0
3 mg/kg	0
10 mg/kg	4
30 mg/kg	5

In the XmAb2513 10mg/kg dose group in 1 mouse the tumor began to regrow on day 43, but 2 days later, on day 45 another mouse no longer had a measurable tumor.

- XmAb2513 was clearly efficient against tumor growth using the L540 (HL) cell line xenograft
- Decreases in rate of tumor growth as well as an increase in survival of the tumor-bearing hosts were observed at all dose levels, and in a dose-related manner

XmAb2513 has comparable binding to cryosections of Human and Cyno tissues

Tissue	Human			Cynomolgus Monkey		
	Stain intensity	# of tissues stained	Frequency of cells stained	Stain intensity	# of tissues stained	Frequency of cells stained
Blood cells-lymphocytes	1+	2/3	v. rare-rare	1-2+	2/3	rare
Bone marrow	1-2+	2/3	rare	1-2+	2/3	rare
Lymph Node	2-3+	**3/3**	v. rare-occas	1-3+	**3/3**	rare-occas
Pancreas	1-2+	1/3	v. rare	1-2+	2/3	v. rare
Skin	2+	1/3	v. rare	2+	1/3	v. rare
Spleen	1-2+	**3/3**	v. rare-rare	1-3+	**3/3**	rare-occas
Tonsil	1-3+	**3/3**	v. rare-occas	1-3+	**3/3**	v. rare-rare

- CD30 antigen has a very limited tissue distribution
- Fluorescein-XmAb2513 was shown to have high affinity for the CD30 antigen
- The binding pattern of fluorescein-XmAb2513 to human tissues was in agreement with reported observations of CD30 expression in human lymphoid tissues – weak to strong staining intensity was present in blood, bone marrow, lymph node, skin, spleen, and tonsil
- The cynomolgus monkey bound fluorescein-XmAb2513 in a similar manner to that of human
- Together with the binding data, the similar staining patterns support the use of the cynomolgus monkey as the appropriate species to characterize XmAb2513 in vivo, including pharmacology, PK, TK, and toxicology studies

XmAb2513 was evaluated in Single Dose and Repeat Dose Cyno Studies

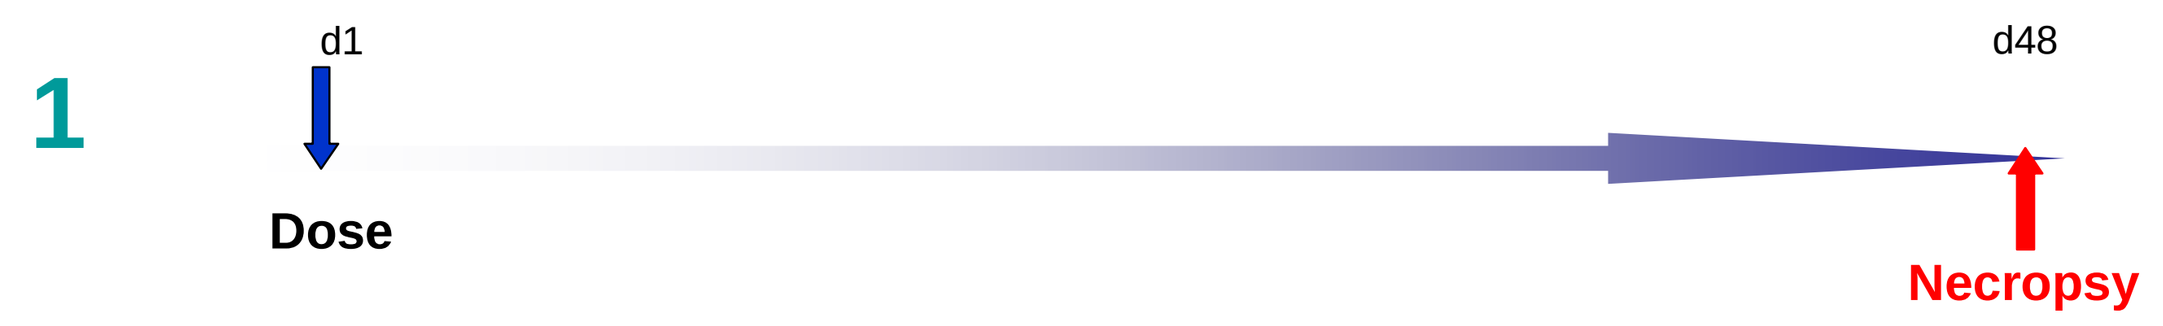
1
d1
d48
Dose
Necropsy

Group No.	No. of Males / Females	Dose Level (mg/kg)	Dose Volume (mL/kg)	Dose Solution Conc. (mg/mL)	Dose Regimen
1	1/1	0 (control)	10	0	1hr iv infusion (0.17 mL/kg/min)
2	1/1	1		0.1	
3	1/1	100		10	

- A single intravenous infusion administration of XmAb2513 at 1 and 100 mg/kg, was well tolerated
- There were no XmAb2513-related effects on clinical observations, body weights, and hematology parameters
- No trends in cytokine levels were observed during the study (IFNγ, IL-1β, IL-2, IL-4, IL-6, IL-8, IL-10, GM-CSF, and TNFα)

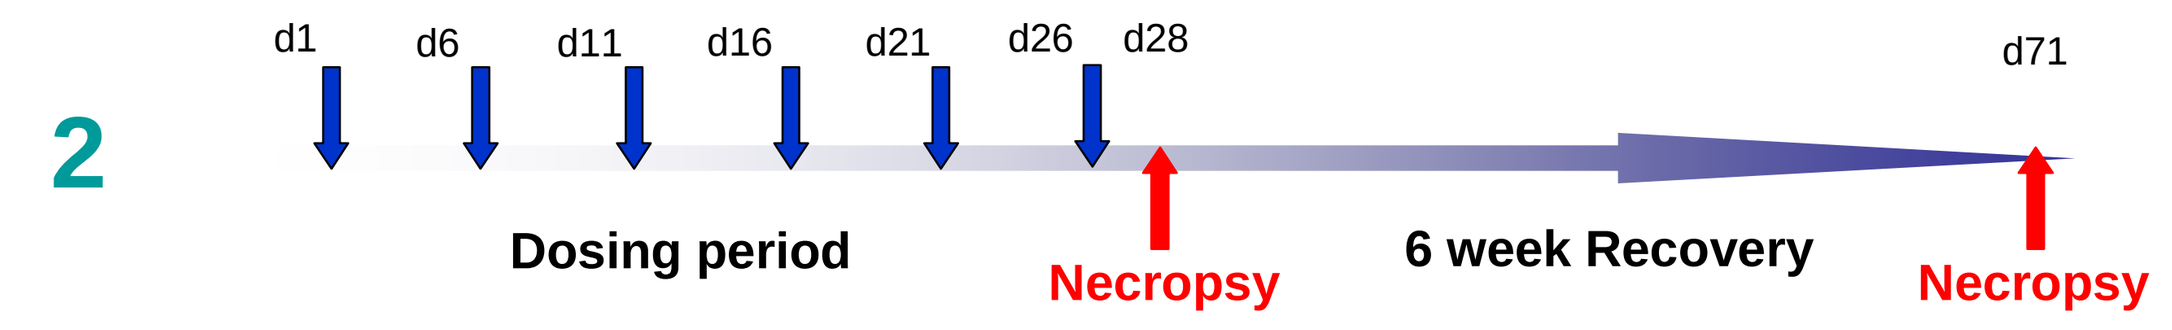
2
d1
d6
d11
d16
d21
d26
d28
d71
Dosing period
Necropsy
6 week Recovery
Necropsy

Group No.	No. of Males / Females	Dose Level (mg/kg)	Dose Volume (mL/kg)	Dose Solution Conc. (mg/mL)	Dose Regimen Q5Dx6	Number Necropsied	
						Day 28	Day 71
1	5/5	0 (control)	10	0	1hr iv infusion (0.17 mL/kg/min)	3/3	2/2
2	3/3	10		1		3/3	--
3	3/3	30		3		3/3	--
4	5/5	100		10		3/3	2/2

- The doses utilized exceeded the maximum proposed clinical dose (12 mg/kg) by either 8.3 fold or 2.67-fold, when compared on a mg/kg,or body surface area (BSA) basis, respectively
- Repeat dose intravenous infusion administration of XmAb2513 at concentrations of 10, 30 and 100 mg/kg was well tolerated as evidenced by in-life observations
- There were no XmAb2513-related effects on clinical observations, body weights, electro-cardiograms, ophthalmic examination parameters, coagulation and urinalysis parameters, or gross observations at necropsy
- XmAb2513-associated changes occurred primarily in the 10 and 30 mg/kg groups and included a reduction in albumin and an increase in globulin serum concentrations, and histologic evidence of hepatic Kupffer cell hypertrophy / hyperplasia, lymph node plasmacytosis, and bone marrow myeloid hypercellularity
- Neutropenia was observed in some of the animals in all dose groups, but was not dose dependent, and was reversible

Repeat dose XmAb2513 effects on Cyno serum neutrophil levels

Vehicle

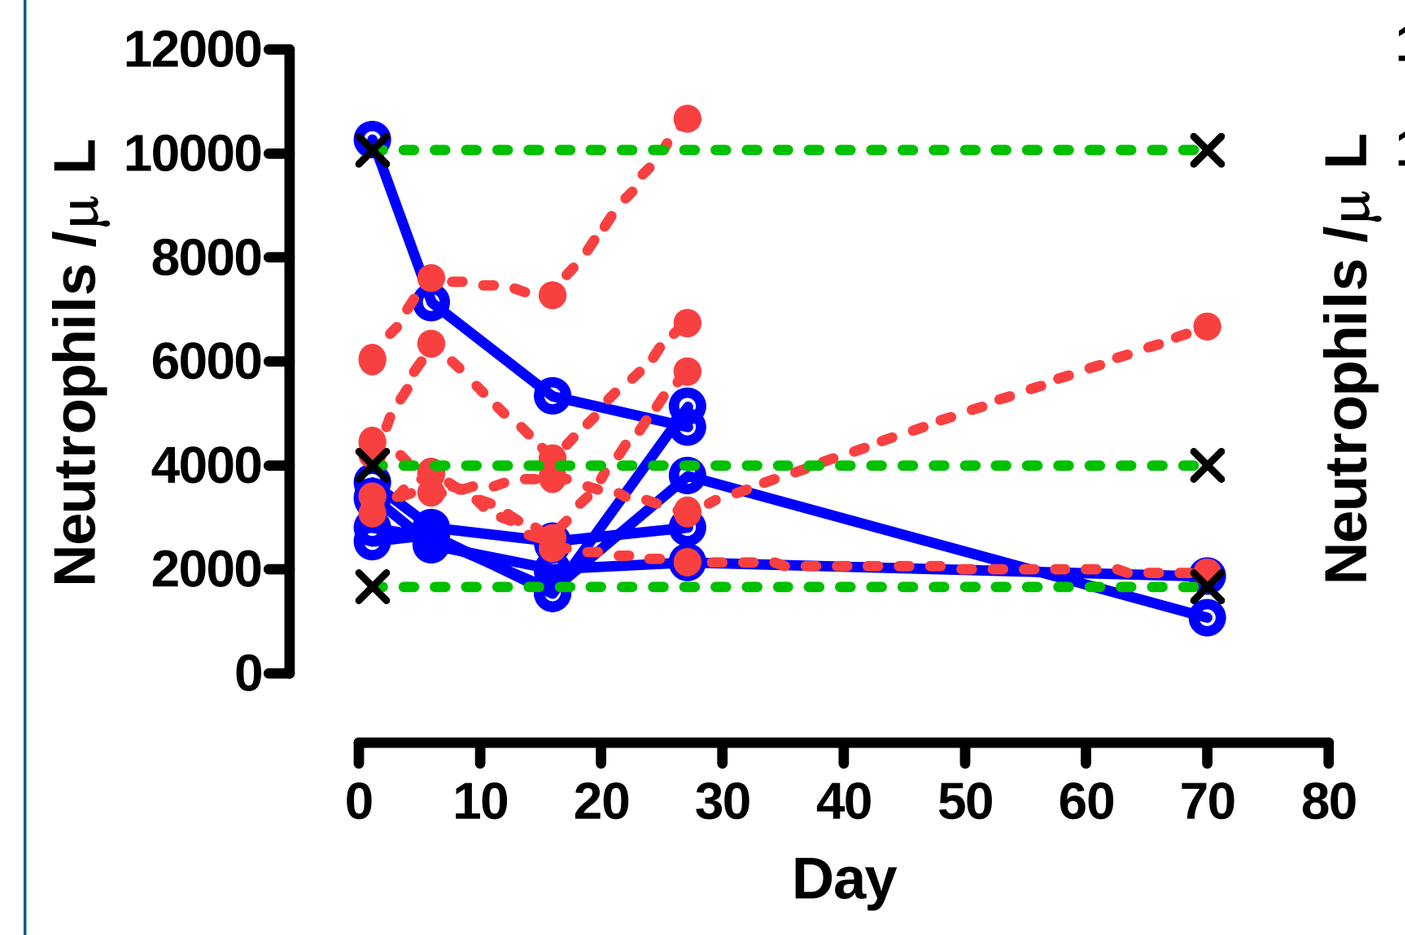
Neutrophils /µ L
Day

100 mg/kg XmAb2513

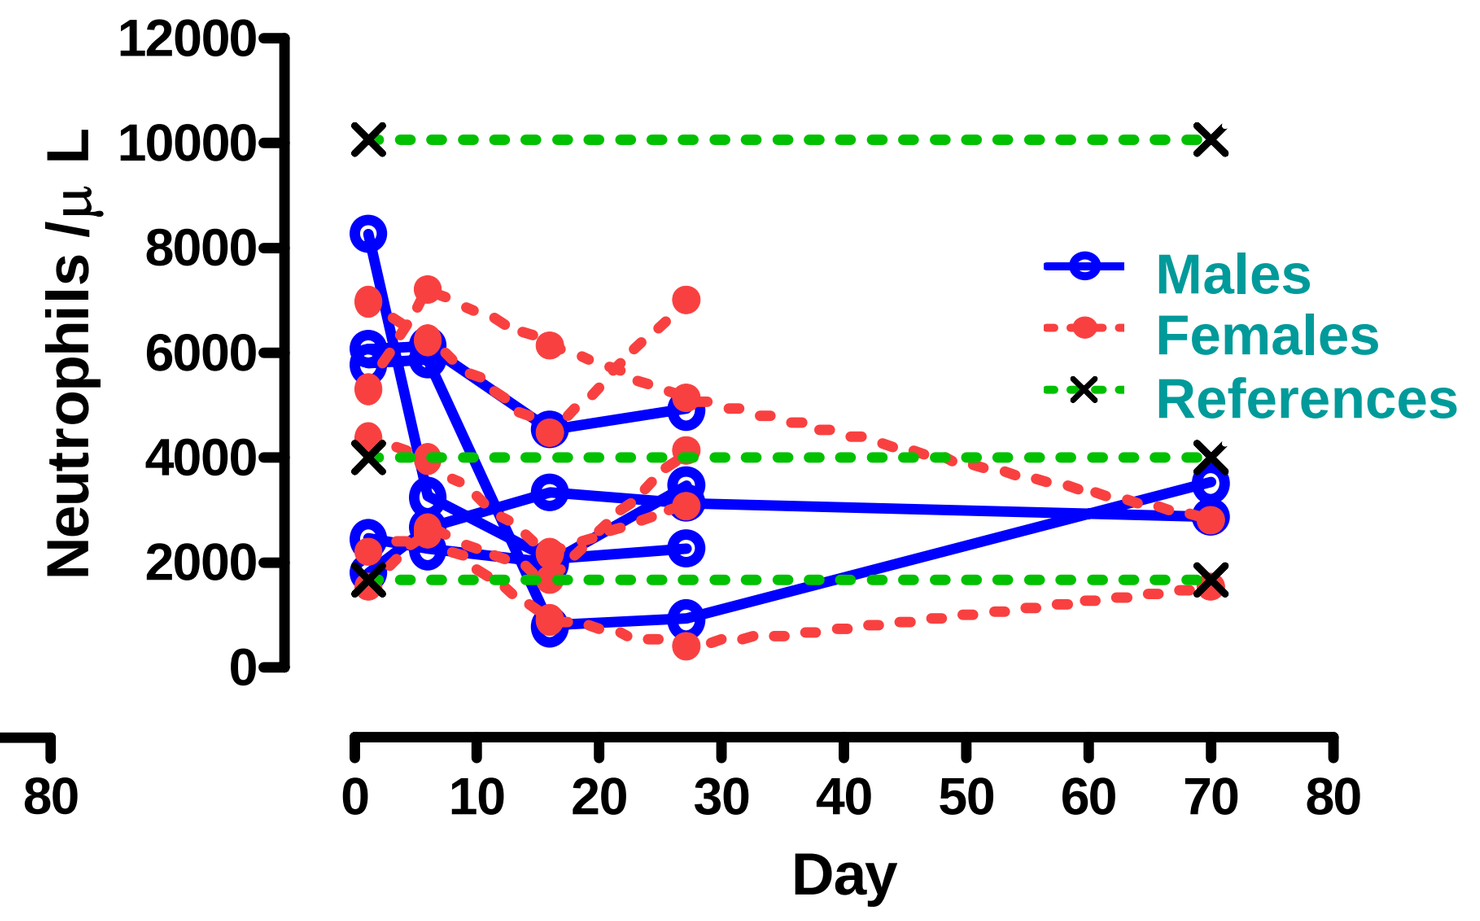
Neutrophils /µ L
Day
Males
Females
References

Exposure and PK in the Cynos after repeat dose XmAb2513

XmAb2513 PK parameters obtained from the Cynomolgus Monkey

Dose Level (mg/kg)	Cmax (µg/mL)	AUC0-t (day * µg/mL)	AUC0-∞ (day * µg/mL)	Half-life t½ (day)	Cl (mL/day /kg)	V1 (mL/kg)	Vss (mL/kg)
10	221	522	1688	17.1	6.14	42.6	126
30	738	1707	4375	13.5	6.86	43.7	115
100	2425	5801	16581	12.6	6.15	42.4	95

AUC0-t is the area under the concentration-time profile from time zero to t days following a single dose
AUC0-∞ is the AUC from time zero to inifinity for a single dose at the specific dose level
Computed using non-compartmental methods

- Non-compartmental analysis was used to determine exposure (Cmax) and (AUC) from all study cynomolgus monkeys
- Exposure to XmAb2513 was proportional to dose (after 1st administration) and increased in a greater than proportional manner with dose (6th administration)
- PK parameters were derived by a 2-compartment model
- Following a rapid initial distribution, the terminal $t_{½}$ for XmAb2513 was between 12.6 to 17.1 days across the three dose levels
- The PK parameters from the cynomolgus monkeys were used to estimate the PK in humans

Mean serum concentrations of XmAb2513 versus time in the Cynomolgus monkeys (actual) and in Humans (modeled) after repeat dose intravenous administrations

Cynos (q5d x6)

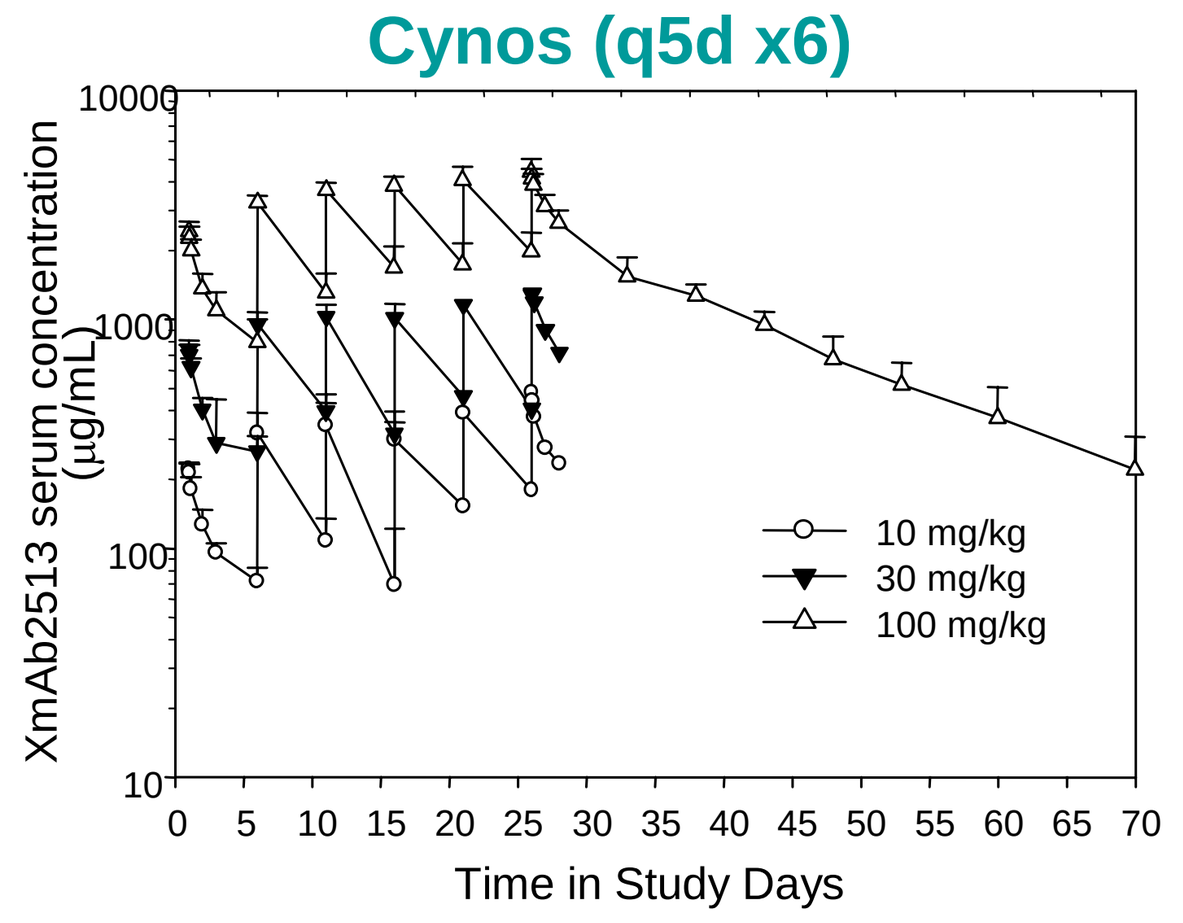
XmAb2513 serum concentration (µg/mL)
10 mg/kg
30 mg/kg
100 mg/kg
Time in Study Days

Humans (qow x4)

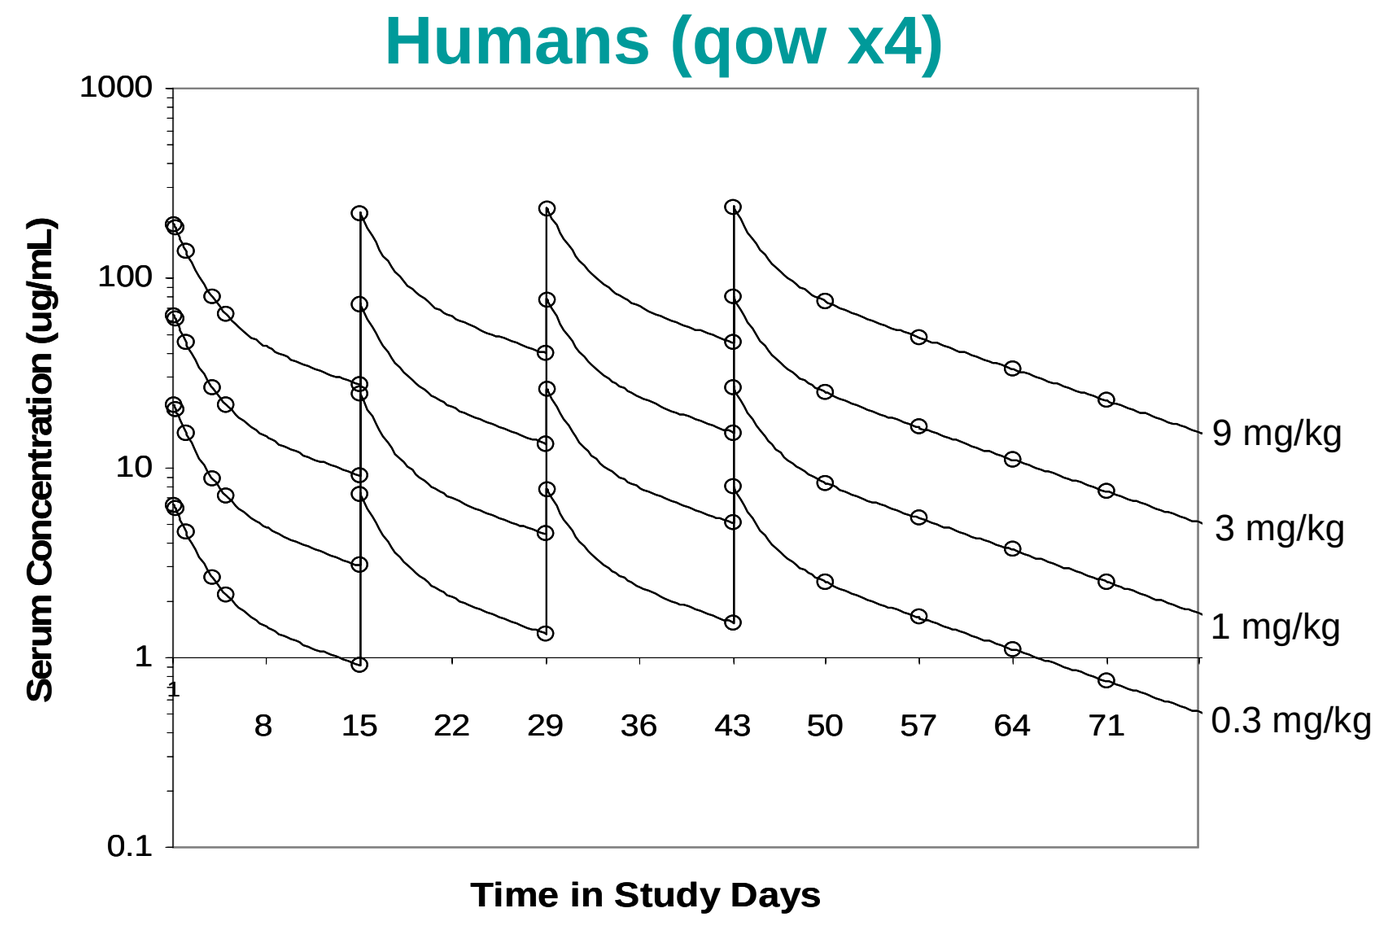
Serum Concentration (ug/mL)
9 mg/kg
3 mg/kg
1 mg/kg
0.3 mg/kg
Time in Study Days

- Based on the safety and PK assessments in the cynomolgus monkey a starting dose of 0.3 mg/kg XmAb2513 is proposed in the clinic
- The terminal $t_{½}$ supports at least an every other week (qow) dosing interval in the clinic
- Modeling showed that a dose level of 0.3 mg/kg XmAb2513 in humans would result in serum trough levels of 1-1.5µg/mL after four infusions given qow
- The clinical starting dose takes into account that the patients enrolled for the planned Phase I trial will be HL or ALCL patients.

Conclusions

- XmAb2513, a novel humanized anti-CD30 mAb, has potent in vitro and in vivo activities
- XmAb2513 is well tolerated in repeat dose cynomolgus toxicity studies
- XmAb2513 will be tested in patients with CD30 positive hematologic malignancies, namely HL and ALCL
- Multiple-dose XmAb2513 administration to patients is supported by the exposure and reasonable toxicity profile observed in cynomolgus monkeys

© 2007 Xencor, Inc., All Rights Reserved, and www.xencor.com